P1303

Impact of Conditioning Intensity on Veno-Occlusive Disease/Sinusoidal Obstruction Syndrome Severity and Outcomes in Adult Hematopoietic Cell Transplant Patients: Results From the DEFIFrance Registry

Mohamad Mohty,[1]* Régis Peffault de Latour,[2] Anne Huynh,[3] Deborah Gutierrez,[4] Nalina Dronamraju,[5] Micha Srour,[6] Didier Blaise[7]

[1]Hôpital St Antoine, Université Sorbonne, INSERM UMRS 938, Paris, France; [2]Hôpital Saint-Louis, Université Paris Cité, Paris, France; [3]Centre Hospitalier Universitaire de Toulouse, Institut Universitaire du Cancer de Toulouse Oncopole, Service d'Hématologie, Toulouse, France; [4]Jazz Pharmaceuticals, Lyon, France; [5]Jazz Pharmaceuticals, Philadelphia, PA, USA; [6]CHU de Lille, Université de Lille, INSERM U1286, INFINITE, Lille, France; [7]Institut Paoli-Calmettes, Aix Marseille Université, Management Sport Cancer Laboratoire, Marseille, France

*Presenting author.

Background

- Veno-occlusive disease/sinusoidal obstruction syndrome (VOD/SOS) is a potentially life-threatening complication of hematopoietic cell transplantation (HCT) and has an associated mortality rate of >80% if left untreated[1,2]
- VOD/SOS occurs due to activation and damage of the sinusoidal endothelium[2,3] and is likely associated with conditioning regimens preceding HCT[4]
- Myeloablative conditioning (MAC) regimens are widely used for patients with hematologic malignancies undergoing autologous and allogeneic HCT, but reduced-intensity conditioning (RIC) regimens have made HCT available for patients with preexisting comorbidities or those unfit to tolerate MAC[5]
- While RIC regimens are thought to decrease the likelihood of VOD/SOS over MAC regimens, this complication does still occur following RIC and can be severe[6,7]
- Defibrotide (Defitelio®) is approved in the European Union for the treatment of severe hepatic VOD/SOS post-HCT in patients aged >1 month.[8] In the United States, defibrotide is approved for the treatment of VOD/SOS in patients with renal or pulmonary dysfunction post-HCT[9]
- Clinical guidelines recommend prompt defibrotide initiation after the diagnosis of VOD/SOS[10,11]
- Defibrotide has been shown to protect the endothelial cells and restore the thrombotic-fibrinolytic balance in vitro[12]
- DEFIFrance was an observational, post-marketing study to collect retrospective and prospective data on patients receiving defibrotide at 53 French transplant centers from July 2014, to March 2020[13]

Objective

- Describe VOD/SOS severity, clinical outcomes, and survival in adult patients who received RIC or MAC regimens followed by defibrotide for the treatment of VOD/SOS after HCT in the DEFIFrance registry

Methods

- Investigators diagnosed VOD/SOS using classical/standard criteria (including but not limited to hyperbilirubinemia, hepatomegaly, ascites, and weight gain >5%)
- In DEFIFrance,[13] VOD/SOS severity was categorized using adult European Society for Blood and Marrow Transplantation (EBMT) criteria for patients aged ⩾18 years
- The primary endpoints were Kaplan-Meier (KM)–estimated day 100 survival and complete response (CR; total serum bilirubin <2 mg/dL and multiorgan failure resolution per investigators' assessment) post-HCT of VOD/SOS at day 100
- A secondary endpoint was incidence of serious treatment-emergent adverse events of special interest (including hemorrhages, coagulopathies, infections, and thromboembolic events)

Results

Table 1. Baseline Patient Demographic and Clinical Characteristics

	Patients Receiving RIC (n=134)	Patients Receiving MAC (n=113)
Median age (range), years	56 (18-74)	47 (18-68)
Male, n (%)	75 (56)	75 (66)
Primary disease, n (%)		
AML	48 (36)	32 (28)
MDS	28 (21)	11 (10)
Lymphoma	18 (13)	32 (28)
ALL	15 (11)	26 (23)
Myelofibrosis	15 (11)	4 (4)
AL mixed phenotype	1 (1)	0 (0)
Other	9 (7)	8 (7)
Number of previous HCTs,[a] n (%)		
0	110 (83)	103 (91)
1	20 (15)	6 (5)
2	2 (1)	4 (4)
3	1 (1)	0
Allogeneic HCT Donor type, n/N (%)	132/134 (99)	93/113 (82)
Non-parent (unrelated)	61/132 (46)	41/93 (44)
HLA similar to siblings	38/132 (29)	33/93 (35)
Haplo-identical	32/132 (24)	16/93 (17)
Umbilical cord blood cells	1/132 (1)	3/93 (3)

[a]Number of prior HCT treatments was missing for 1 patient receiving RIC and was excluded from percentage calculations.
AL, acute leukemia; ALL, acute lymphoblastic leukemia; AML, acute myeloid leukemia; HCT, hematopoietic cell transplantation; HLA, human leukocyte antigen; MAC, myeloablative conditioning; MDS, myelodysplastic syndrome; RIC, reduced-intensity conditioning.

- The median (interquartile range [IQR]) time from HCT to VOD/SOS diagnosis was shorter in the RIC subgroup (12 [6, 21] days) vs the MAC subgroup (14 [9, 26] days)
- The median (IQR) duration of defibrotide treatment was similar for both subgroups (RIC: 15 [10, 22] days; MAC: 16 [12, 22] days)

Figure 1. Severity of VOD/SOS for Patients Treated With RIC or MAC

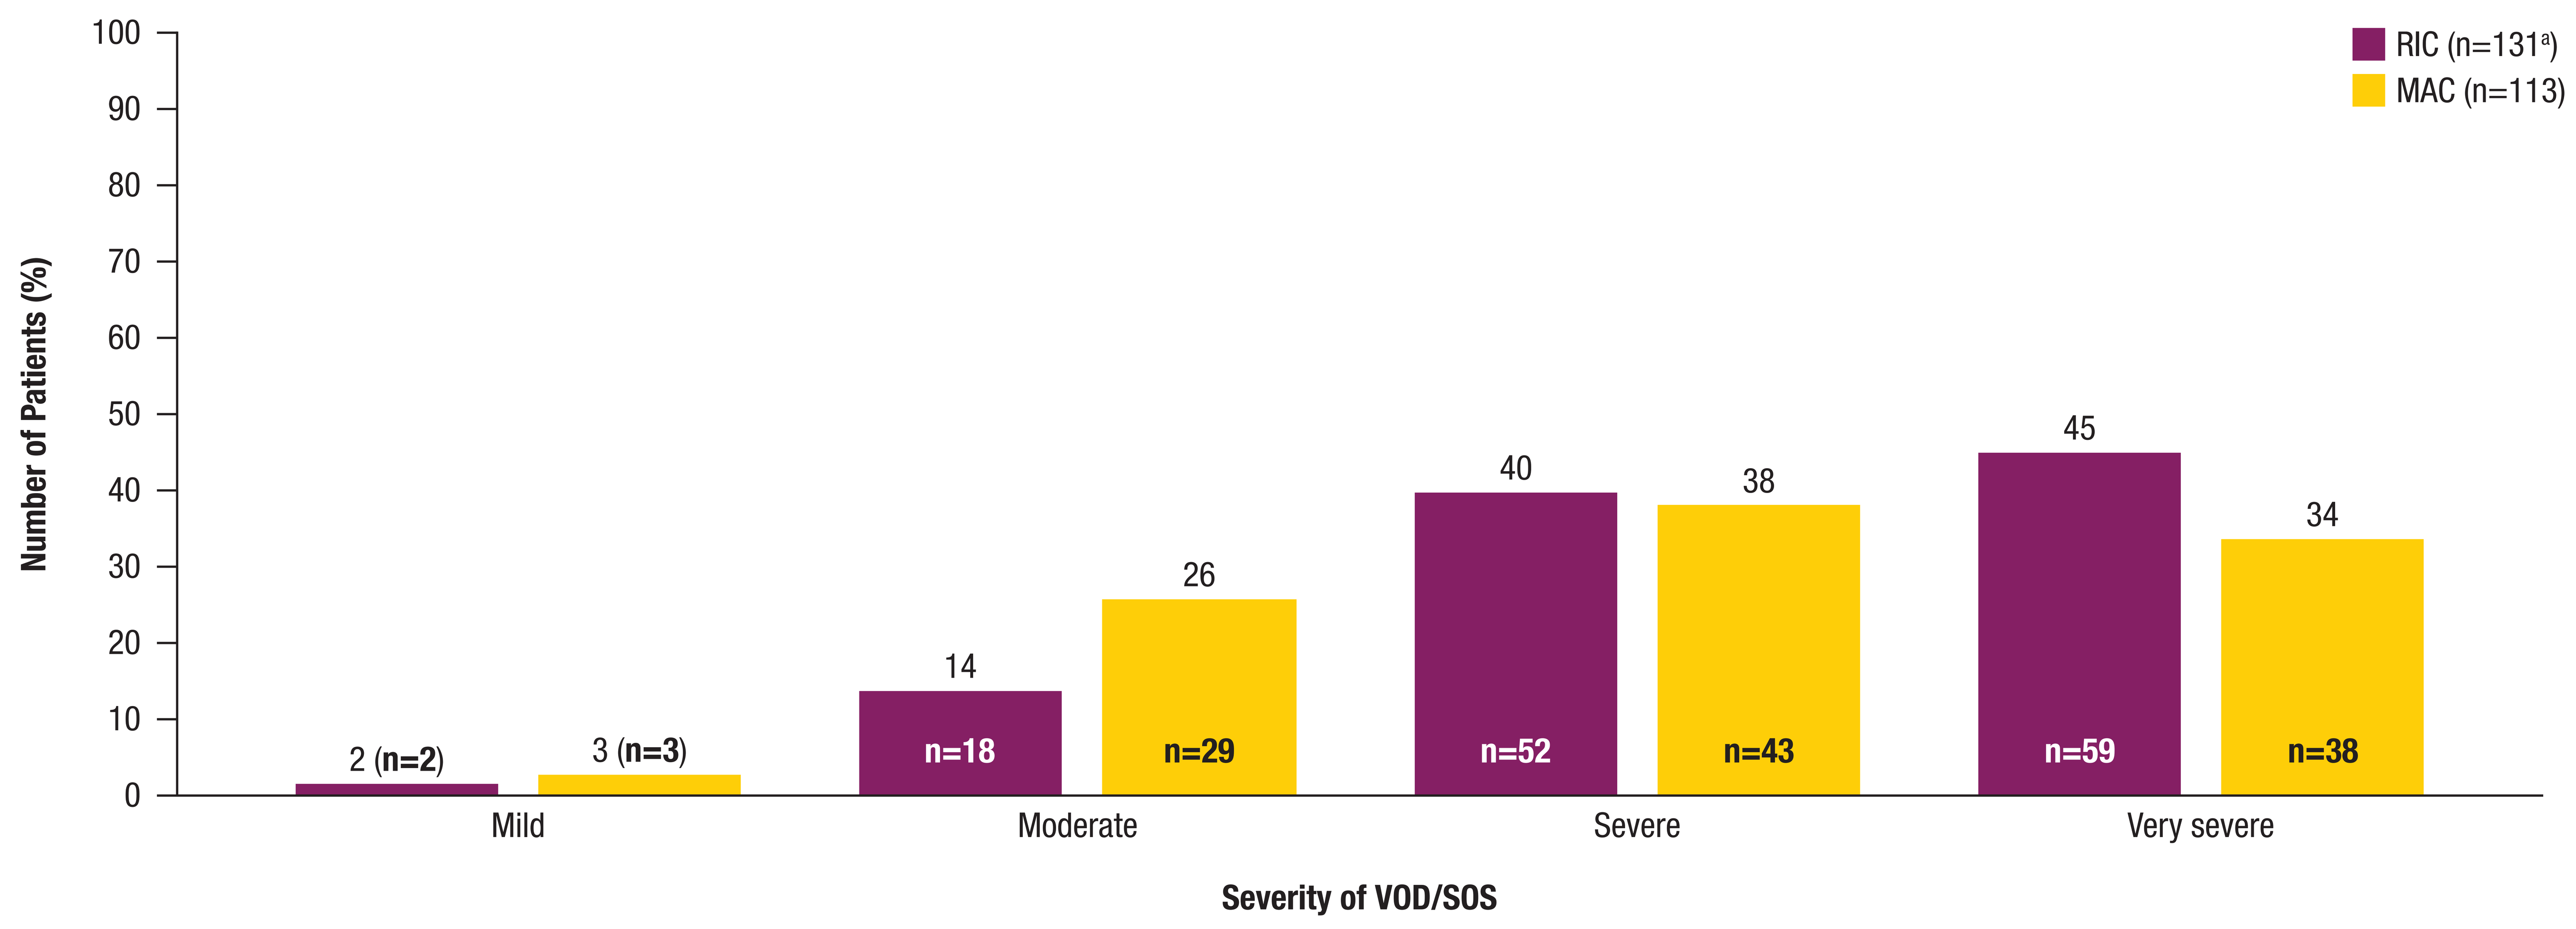

RIC (n=131[a])
MAC (n=113)
Number of Patients (%)
100
90
80
70
60
50
40
30
20
10
0
2 (n=2)
3 (n=3)
14
n=18
26
n=29
40
n=52
38
n=43
45
n=59
34
n=38
Mild
Moderate
Severe
Very severe
Severity of VOD/SOS

[a]VOD/SOS severity was missing for 3 patients receiving RIC and were excluded from percentage calculations.
MAC, myeloablative conditioning; RIC, reduced-intensity conditioning; SOS, sinusoidal obstruction syndrome; VOD, veno-occlusive disease.

- A higher proportion of very severe VOD/SOS was noted in patients receiving RIC (45%) vs MAC (34%), with a corresponding lower proportion of moderate VOD/SOS (14% vs 26%)

Figure 2. KM-Estimated Day 100 and 1 Year Post-HCT Survival of Patients Receiving RIC or MAC

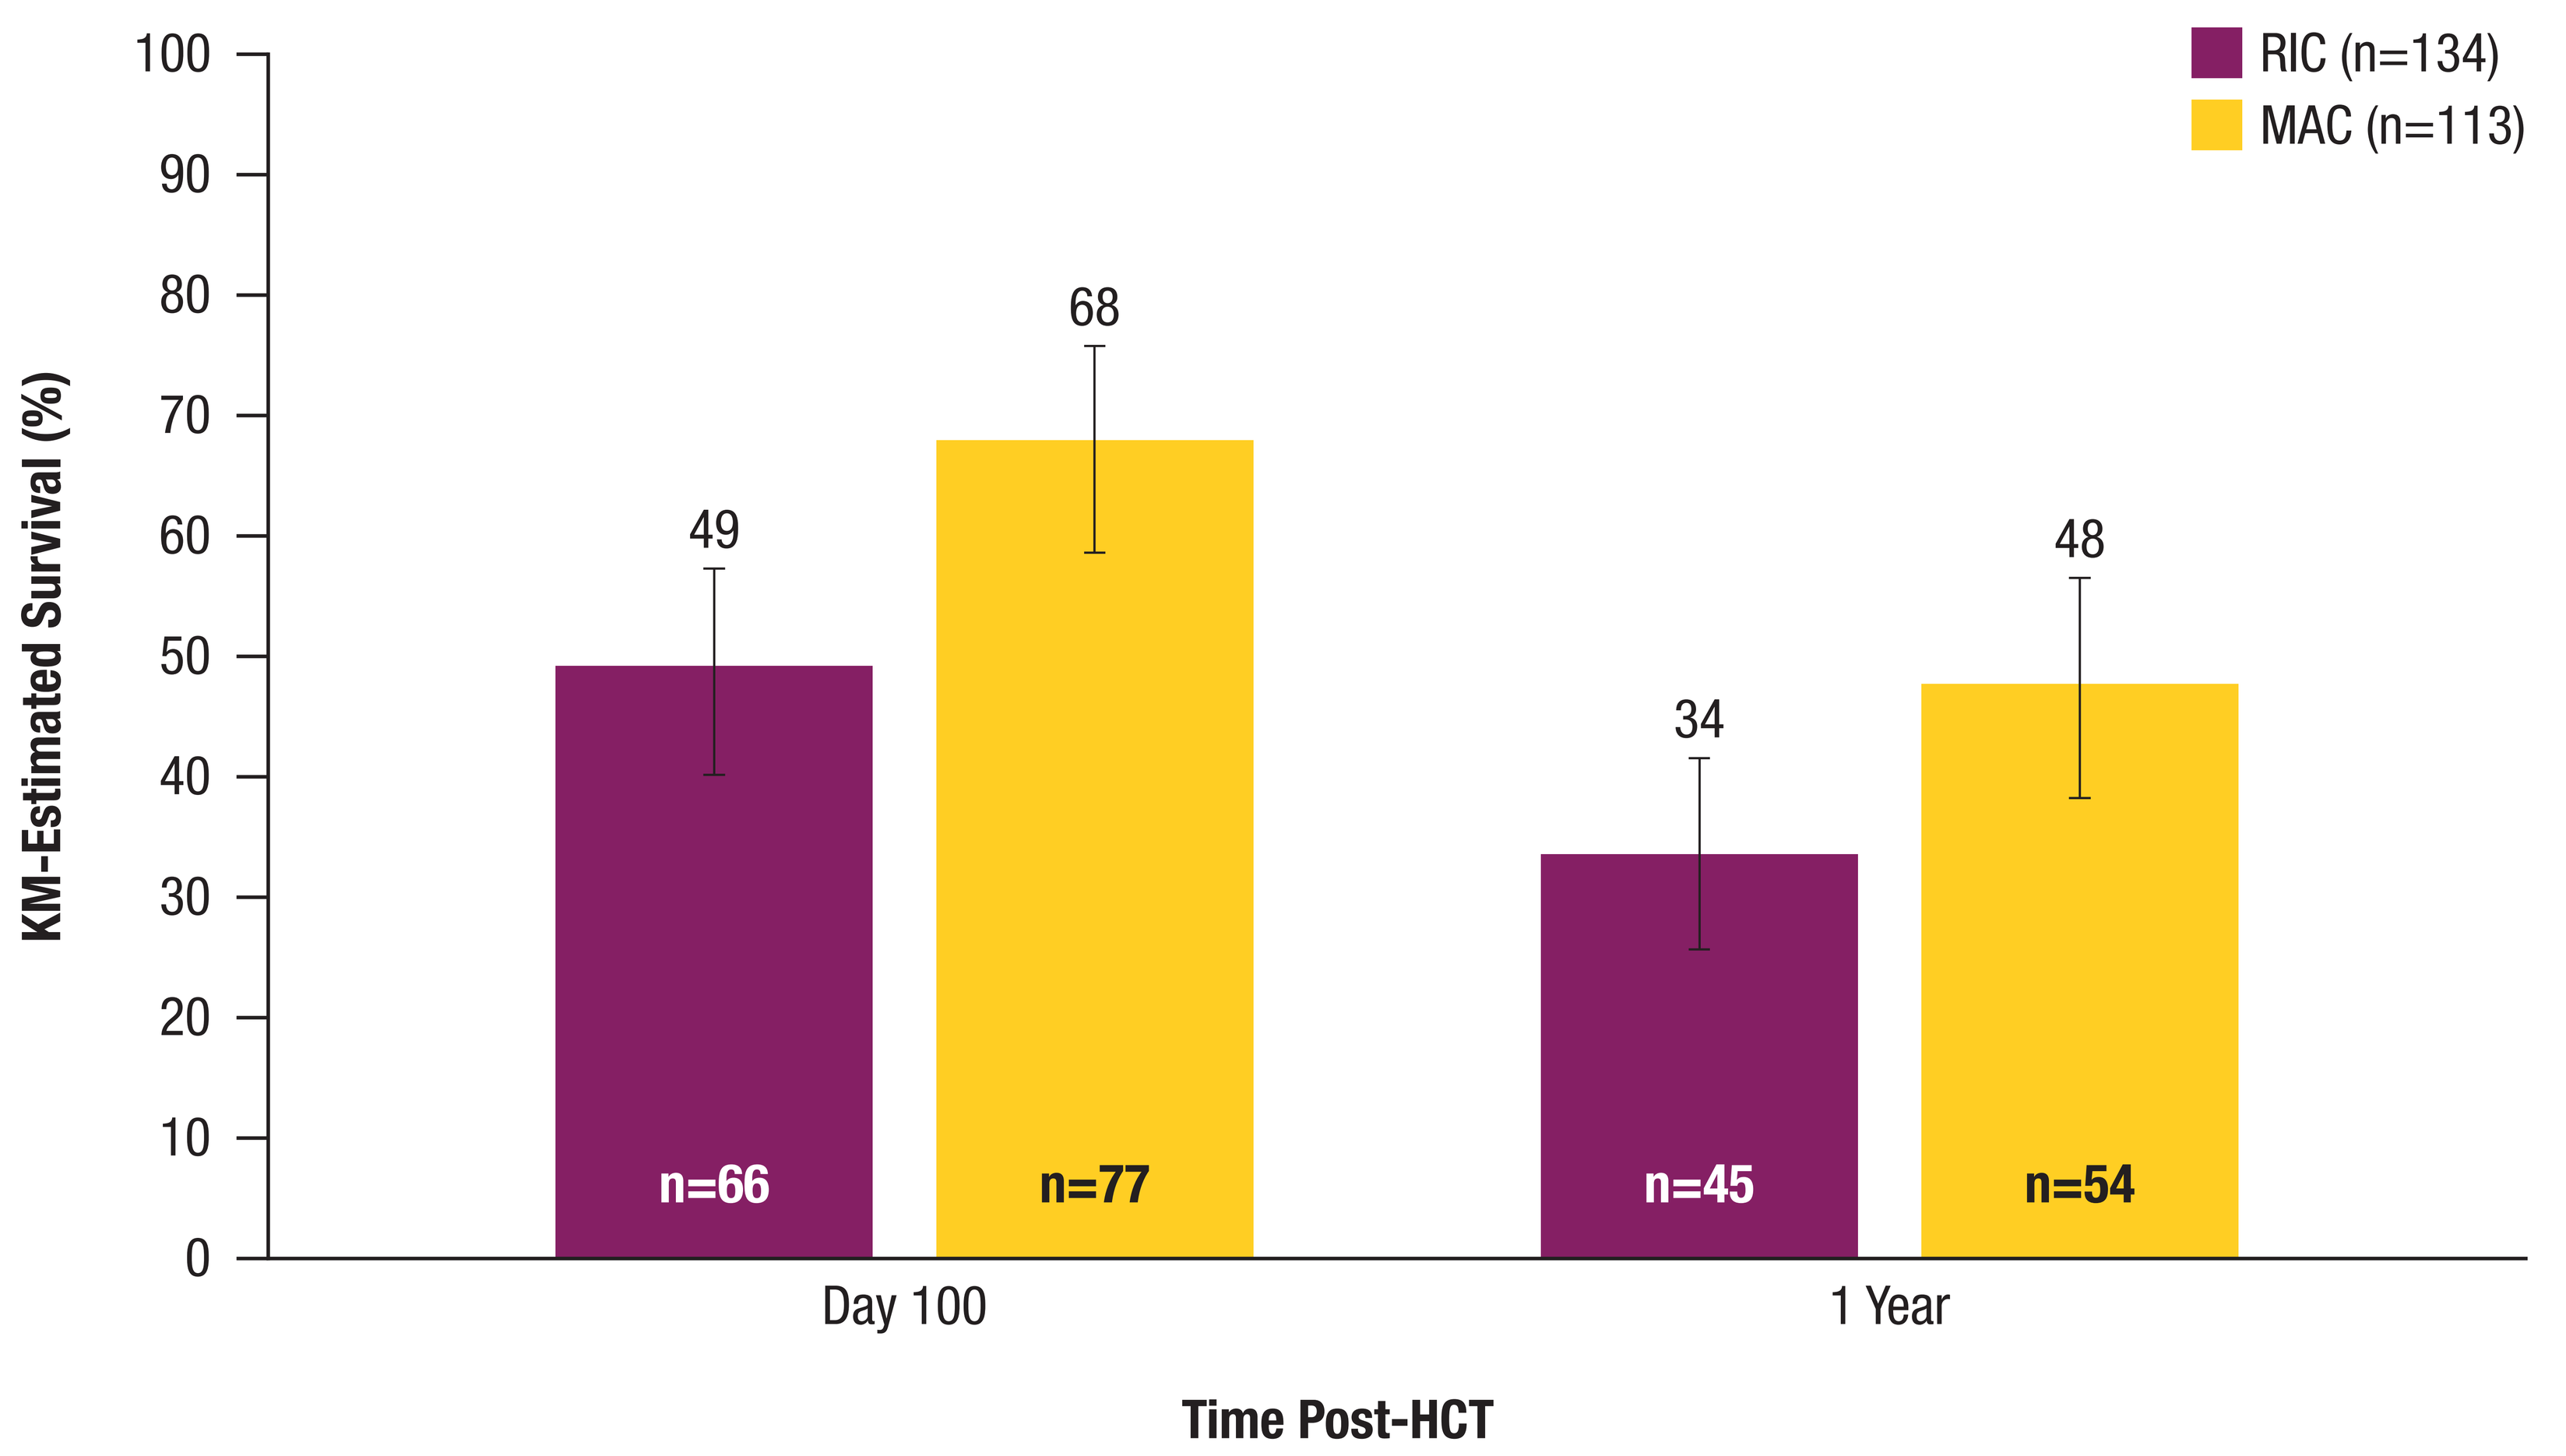

RIC (n=134)
MAC (n=113)
KM-Estimated Survival (%)
100
90
80
70
60
50
40
30
20
10
0
49
n=66
68
n=77
34
n=45
48
n=54
Day 100
1 Year
Time Post-HCT

Whiskers are 95% CI.
CI, confidence interval; HCT, hematopoietic cell transplantation; KM, Kaplan-Meier; MAC, myeloablative conditioning; RIC, reduced-intensity conditioning.

- KM-estimated day 100 post-HCT survival was 49% (95% confidence interval [CI]: 40%, 57%) in patients receiving RIC and 68% (95% CI: 59%, 76%) in patients receiving MAC
- KM-estimated 1-year post-HCT survival was 34% (95% CI: 26%, 42%) in patients receiving RIC and 48% (95% CI: 38%, 57%) in patients receiving MAC

Figure 3. Day 100 Post-HCT CR of Patients Receiving RIC or MAC

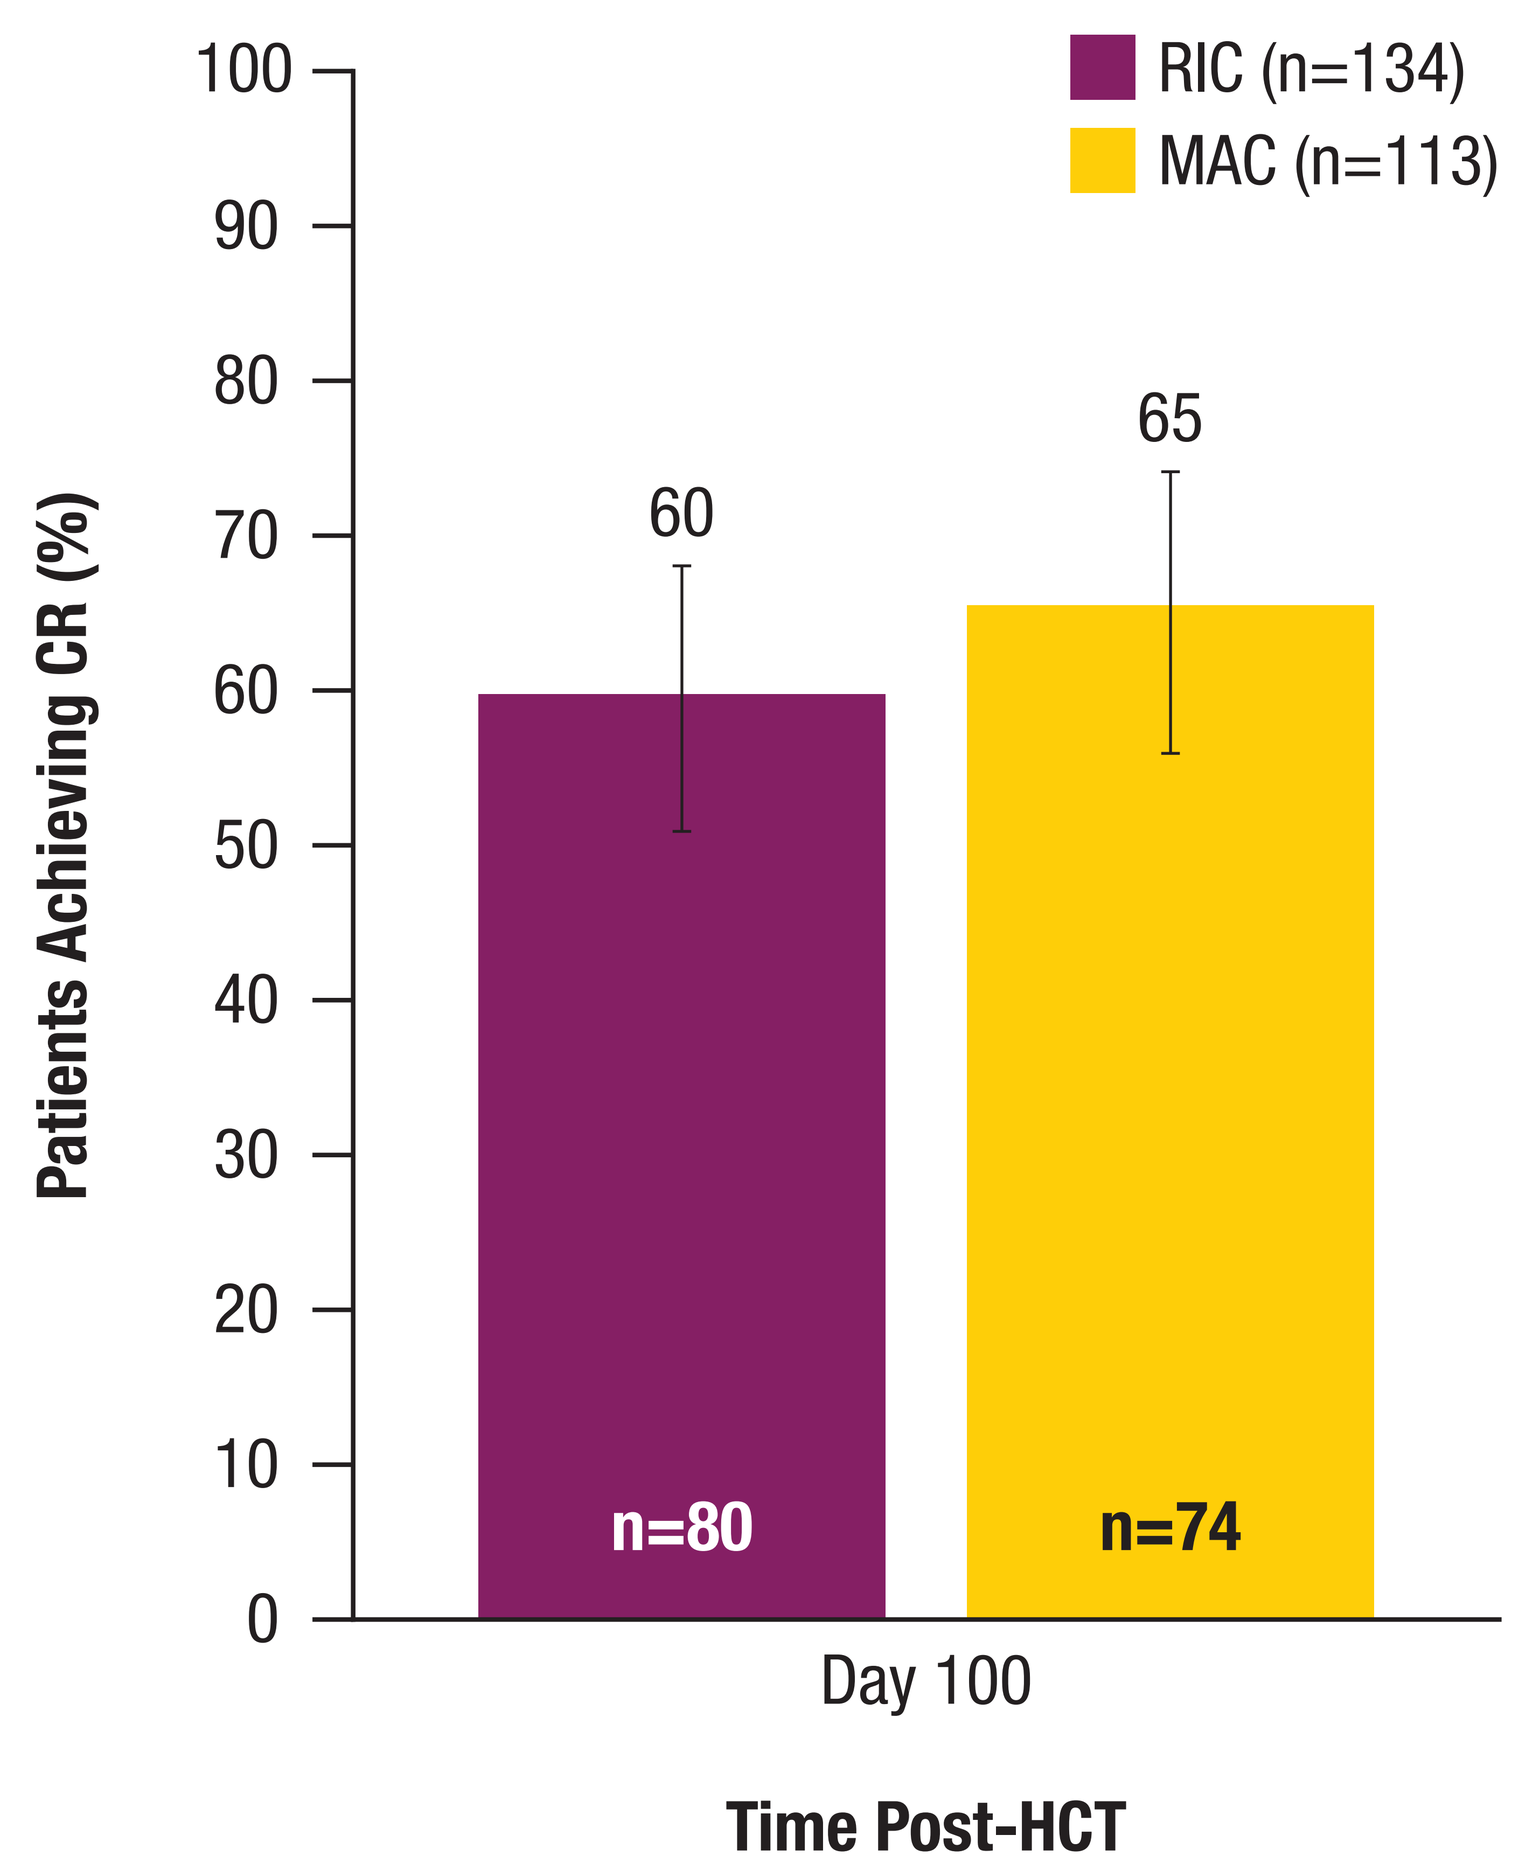

RIC (n=134)
MAC (n=113)
Patients Achieving CR (%)
100
90
80
70
60
50
40
30
20
10
0
60
n=80
65
n=74
Day 100
Time Post-HCT

Whiskers are 95% CI.
CI, confidence interval; CR, complete response; HCT, hematopoietic cell transplantation; MAC, myeloablative conditioning; RIC, reduced-intensity conditioning.

Table 2. Serious TEAEs of Special Interest

Variable	Patients Receiving RIC (n=134)	Patients Receiving MAC (n=113)
Any Serious TEAEs of Special Interest, n (%)	45 (34)	38 (34)
Infections	29 (22)	14 (12)
Hemorrhages	23 (17)	25 (22)
Coagulopathies	2 (1)	3 (3)
Thromboembolic events	0	2 (2)

MAC, myeloablative conditioning; RIC, reduced-intensity conditioning; TEAE, treatment-emergent adverse event.

- Serious treatment-emergent adverse events of special interest were reported in 34% of patients in each subgroup
- Overall, transplant-related mortality by 1 year was 55% in patients treated with RIC (n=74) and 41% in patients treated with MAC (n=46)
- Of all patient deaths, the majority were related to HCT, both for patients treated with RIC (n=74/88, 84%) and MAC (n=46/59, 78%). These included infection (RIC: n=39, 44%; MAC: n=17, 29%) and VOD/SOS (RIC: n=20, 23%; MAC: n=17, 29%)

Conclusions

- Adult patients who receive RIC regimens are still at risk of VOD/SOS, which may be related to characteristics such as older age and previous HCT[4]
 - 45% of patients who received RIC in this analysis had very severe VOD/SOS
- A substantial proportion of patients receiving defibrotide achieved complete resolution of VOD/SOS symptoms in both RIC- and MAC-conditioned groups
- Differences in patient characteristics between those treated with RIC vs MAC, including age, primary disease diagnosis, and donor type, may contribute to differences in VOD/SOS severity and survival
- These results highlight the need to maintain vigilance for VOD/SOS signs and symptoms following RIC, as prompt diagnosis of VOD/SOS may improve outcomes

References: 1. Coppell JA, et al. *Biol Blood Marrow Transplant.* 2010;16(2):157-168. **2.** Richardson PG, et al. *Blood Adv.* 2018;2(12):1495-1509. **3.** Richardson PG, et al. *Expert Opin Drug Saf.* 2013;12(1):123-136. **4.** Putta S, et al. *Transplant Cell Ther.* 2023;29(3):166.e1-166.e10. **5.** Gyurkocza B, et al. *Blood.* 2014;124(3):344-353. **6.** Lewis C, et al. *Biol Blood Marrow Transplant.* 2020;26(3):529-539. **7.** Tsirigotis PD, et al. *Bone Marrow Transplant.* 2014;49(11):1389-1392. **8.** Jazz Pharmaceuticals Ireland, Limited. Defitelio® (defibrotide) Summary of Product Characteristics, 2023. Available at: https://www.ema.europa.eu/en/documents/product-information/defitelio-epar-product-information_en.pdf. Last accessed January 22, 2024. **9.** Jazz Pharmaceuticals, Inc. Defitelio® (defibrotide) Prescribing Information, 2022. Available at: https://pp.jazzpharma.com/pi/defitelio.en.USPI.pdf. Last accessed January 22, 2024. **10.** Mahadeo KM, et al. *Lancet Haematol.* 2020;7(1):e61-e72. **11.** Kernan NA, et al. *Br J Haematol.* 2018;181(6):816-827. **12.** Palomo M, et al. *Biol Blood Marrow Transplant.* 2011;17(4):497-506. **13.** Mohty M, et al. *Bone Marrow Transplant.* 2023;58(4):367-376.

Support and Acknowledgments: This study was supported by Jazz Pharmaceuticals. Medical writing support, under the direction of the authors, was provided by Sarah Fauque, PhD, of CMC Affinity, a division of IPG Health Medical Communications, with funding from Jazz Pharmaceuticals, in accordance with Good Publication Practice (GPP 2022) guidelines.

Disclosures: M Mohty has received research funding and honoraria from Jazz Pharmaceuticals. **R Peffault de Latour**, **A Huynh**, and **D Blaise** have received honoraria from Jazz Pharmaceuticals. **D Gutierrez** and **N Dronamraju** are employees of Jazz Pharmaceuticals and hold stock and/or stock options in Jazz Pharmaceuticals. **M Srour** has no disclosures to declare.

Copies of this poster obtained through Quick Response code are for personal use only and may not be reproduced without permission from EHA and the authors of this poster.

Poster presented at the EHA 2024 (European Hematology Association) Hybrid Congress, June 13–16, 2024, Madrid, Spain.