LB-A021

Zanidatamab in patients with advanced HER2-positive salivary gland cancer: A pooled analysis of early-phase studies

Keun-Wook Lee[1], Elena Elimova[2], Do-Youn Oh[3], Muralidhar Beeram[4], Toshihiko Doi[5], Kay T Yeung[6], Theresa Samuel Nached[7], Kavita V Shah[7], Douglas S Fuller[8], Funda Meric-Bernstam[9]*

*Presenting author

[1]Seoul National University College of Medicine, Seoul National University Bundang Hospital, Seongnam, Republic of Korea; [2]Princess Margaret Cancer Centre, Toronto, ON, Canada; [3]Seoul National University Hospital, Cancer Research Institute, Seoul National University College of Medicine, Seoul, Republic of Korea; [4]START San Antonio, San Antonio, TX, USA; [5]National Cancer Center Hospital East, Kashiwa, Japan; [6]University of California San Diego, Moores Cancer Center, La Jolla, CA, USA; [7]Jazz Pharmaceuticals, Palo Alto, CA, USA; [8]Jazz Pharmaceuticals, Philadelphia, PA, USA; [9]The University of Texas MD Anderson Cancer Center, Houston, TX, USA

Plain Language Summary

Why did we perform this research?

- People with advanced salivary gland cancer that has too much of a protein called HER2 often have a short life expectancy and limited treatment options

How did we perform this research?

- This analysis looked at whether a medicine called zanidatamab, which targets HER2, could help patients with salivary gland cancer, a disease for which there is no established treatment option

What were the results of this research?

- This initial analysis showed that these patients may benefit from zanidatamab
- All patients who received treatment had their tumors shrink and survived for many months
- Zanidatamab-related side effects were manageable and did not cause any of the patients to stop treatment

Background

- Advanced salivary gland cancers (SGC) are rare and heterogeneous tumors that are associated with poor outcomes[1,2]
- There are no established chemotherapy or targeted treatment regimens, highlighting the need for personalized treatment strategies guided by actionable molecular targets[1]
- Human epidermal growth factor receptor 2 (HER2) is often overexpressed in SGCs, affecting up to 43% of patients depending on the histological subtype[3]
- Zanidatamab is a dual HER2-targeted bispecific antibody that binds to 2 distinct sites on HER2 in a *trans* configuration, driving HER2 crosslinking not observed with trastuzumab or trastuzumab + pertuzumab[4]
 - It has multiple antitumor mechanisms of action, including reduction of HER2 signaling and activation of immune-mediated effects (complement-dependent cytotoxicity as well as antibody-dependent cellular cytotoxicity and phagocytosis)
- Zanidatamab has received accelerated approval in the US and conditional approvals in China and the EU for previously treated, advanced HER2 immunohistochemistry (IHC) 3+ biliary tract cancer[5]
- In the first-in-human (FIH) phase 1 study, zanidatamab demonstrated preliminary antitumor activity in 11 different types of HER2-expressing solid tumors, including SGC[6]

Figure 1. Zanidatamab Structure and Targeted Binding[4]

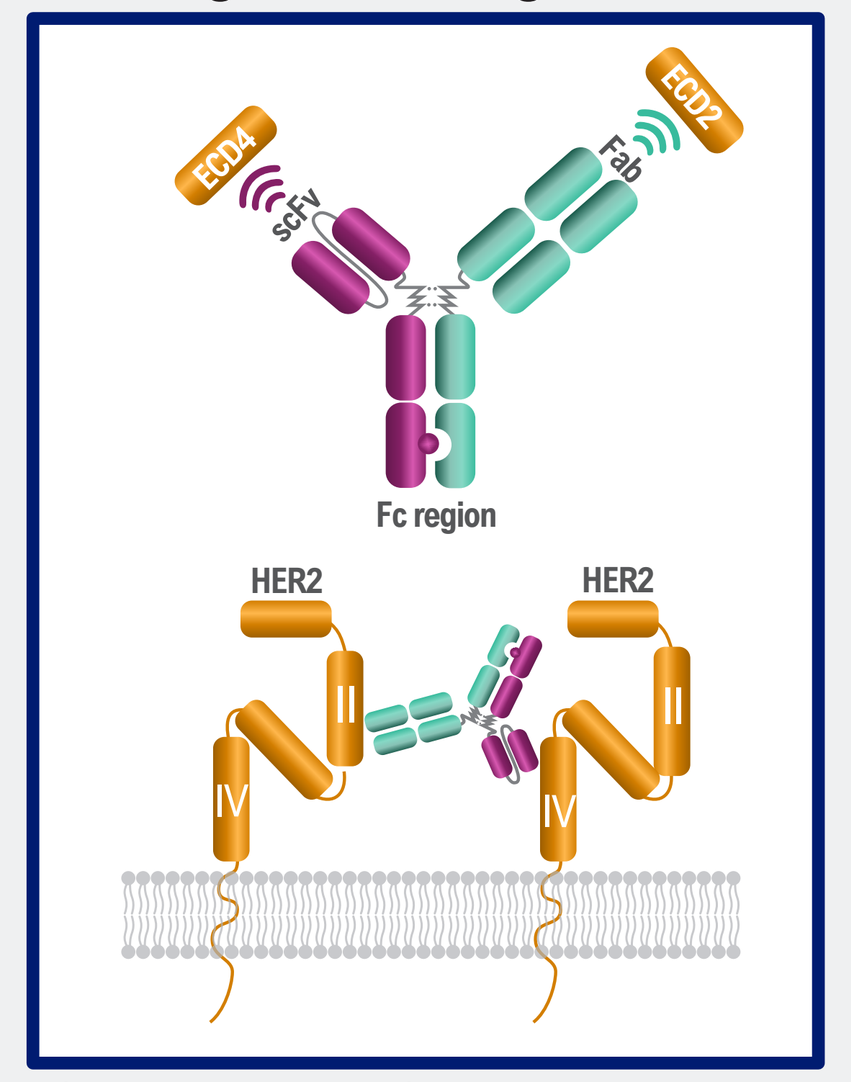

ECD4
scFv
ECD2
Fab
Fc region
HER2
HER2
II
IV

ECD, extracellular domain; Fab, fragment antigen-binding; Fc, fragment crystallizable; HER2, human epidermal growth factor receptor 2; scFv, single-chain variable fragment.

Objective

- To evaluate the safety and efficacy of zanidatamab in patients with heavily pretreated, advanced HER2-positive SGCs from 3 early-phase clinical trials

Methods

- Adult patients with previously treated, unresectable locally advanced or metastatic HER2-expressing SGC were enrolled in 3 zanidatamab trials
 - FIH phase 1 study (ZW25-101; NCT02892123; data cutoff September 30, 2024)
 - Phase 1 study in Japan (study ZW25-102; JRCT2031210161; data cutoff June 23, 2025)
 - Phase 1b/2 study of zanidatamab + evorpacept, a high-affinity CD47 inhibitor (study ZW25-204; NCT05027139; data cutoff June 2, 2025)
- Primary objectives of the FIH and ZW25-102 studies were to characterize the safety and tolerability of zanidatamab monotherapy
- Primary objectives of study ZW25-204 were to assess the safety, tolerability, and antitumor activity of zanidatamab + evorpacept
- Patients who received zanidatamab monotherapy were pooled for summary analysis

Figure 2. Study Details for Patients With SGC From Early-Phase Zanidatamab Trials

Eligibility criteria across trials
- Locally advanced and/or metastatic solid tumors, including SGC, that progressed after all known efficacious therapies

FIH
- Adults (aged ≥18 years)
- HER2-positive (IHC 3+ or IHC 2+/FISH+)

ZW25-102
- Japanese adults (aged ≥20 years)
- HER2-expressing (IHC 1+, 2+, or 3+)

ZW25-204
- Adults (aged ≥18 years)
- HER2-positive (IHC 3+ or IHC 2+/FISH+)

Zanidatamab administration across trials
- Patients received mandatory prophylaxis[a] for potential infusion-related reactions before every zanidatamab infusion

FIH
- 20 mg/kg IV Q2W
- 28-day treatment cycle

ZW25-102
- 1800 mg (patients <70 kg) IV Q3W
- 21-day treatment cycle

ZW25-204
- 1600 mg (patients ≥70 kg) IV Q2W
- Evorpacept 30 mg/kg IV Q2W
- 28-day treatment cycles

CT/MRI scans Q8W per RECIST v1.1

Select endpoints across trials
- **Safety and tolerability**
 - AEs and serious AEs
 - Laboratory abnormalities
 - Dose reductions
- **Antitumor activity**
 - Investigator-assessed cORR
 - DCR
 - DOR
 - PFS

[a]Acetaminophen, antihistamines, and corticosteroids.
AE, adverse event; cORR, confirmed objective response rate; CT, computed tomography; DCR, disease control rate; DOR, duration of response; FIH, first-in-human; FISH, fluorescence in situ hybridization; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; IV, intravenous; MRI, magnetic resonance imaging; PFS, progression-free survival; Q2W, every 2 weeks; Q3W, every 3 weeks; Q8W, every 8 weeks; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; SGC, salivary gland cancer.

Results

Table 1. Baseline Demographics and Clinical Characteristics of Patients From 3 Early-Phase Trials

Pt	Study	Zanidatamab dose	Age	Sex	Race or nationality	ECOG PS	Histology	Prior brain mets	HER2 status[a]	Prior HER2-targeted therapy
1	FIH	20 mg/kg Q2W	75	M	White	1	SDC	No	IHC 2+/ FISH+	Trastuzumab + pertuzumab
2	FIH	20 mg/kg Q2W	63	M	Asian	1	SDC	No	IHC 3+/ FISH+	Trastuzumab + pertuzumab
3	FIH	20 mg/kg Q2W	52	F	Asian	0	SDC	Yes	IHC 3+/ FISH+	None
4	FIH	20 mg/kg Q2W	69	M	Asian	1	SDC	Yes	IHC 3+/ FISH+	Trastuzumab
5	FIH	20 mg/kg Q2W	68	M	Asian	1	SDC	Yes	IHC 3+/ FISH+	Trastuzumab
6	FIH	20 mg/kg Q2W	65	M	Asian	0	SDC	No	IHC 3+/ FISH+	Trastuzumab
7	FIH	20 mg/kg Q2W	52	F	Asian	0	SDC	No	IHC 3+/ FISH+	None
8	FIH	20 mg/kg Q2W	71	M	White	1	Adenocarcinoma	No	IHC 3+/ FISH+	None
9	ZW25-102	1800 mg Q3W	69	M	Japanese	1	Adenocarcinoma	No	IHC 3+/ FISH+	None
10	ZW25-204	1600 mg Q2W + evorpacept	55	M	Iranian	1	SDC, ex-pleomorphic adenoma	No	IHC 3+/ FISH+	Trastuzumab + pertuzumab; T-DM1

[a]Based on central laboratory results or local laboratory results if central results were not available.
ECOG PS, Eastern Cooperative Oncology Group performance status; F, female; FIH, first-in-human; FISH, fluorescence in situ hybridization; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; M, male; mets, metastasis; Pt, patient; Q2W, every 2 weeks; Q3W, every 3 weeks; SDC, salivary duct carcinoma; T-DM1, trastuzumab emtansine.

- Nine patients with advanced SGC received zanidatamab monotherapy (FIH, n = 8; study ZW25-102, n = 1), and 1 patient received zanidatamab in combination with evorpacept (study ZW25-204)
- Five (56%) patients who received monotherapy had prior HER2-targeted therapy (trastuzumab, n = 3; trastuzumab + pertuzumab, n = 2), and the patient who received combination therapy had prior HER2-targeted therapies (trastuzumab + pertuzumab and trastuzumab emtansine)
- Among patients who received zanidatamab monotherapy,
 - The median (range) age was 68 (52–75) years
 - Most patients (7 [78%]) were male
 - The median (range) duration of follow-up was 10.0 (4.1–56.9) months
 - The median (range) duration of treatment was 8.8 (3.5–56.3) months

Table 2. Pooled Safety Summary of Patients Receiving Zanidatamab Monotherapy

n (%)	Salivary gland cancer (n = 9)
Any AE	9 (100)
Grade 3[a]	2 (22)
Serious	1 (11)
Leading to dose reduction	0
Leading to zanidatamab discontinuation	1 (11)
Any TRAE	7 (78)
Grade ≥3	0
Serious	0
Leading to dose reduction	0
Leading to zanidatamab discontinuation	0
Grade 1/2 TRAEs occurring in ≥20% of patients	
Infusion-related reaction	4 (44)
Diarrhea	4 (44)
Fatigue	2 (22)
Rash	2 (22)

[a]There were no grade >3 AEs.
AE, adverse event; TRAE, treatment-related AE.

- One patient discontinued zanidatamab due to a serious, grade 3 pulmonary embolism that was not considered related to treatment
- No serious or grade ≥3 TRAEs occurred in patients receiving zanidatamab monotherapy
- The patient who received evorpacept + zanidatamab did not experience any grade ≥3 AEs, serious AEs, or AEs that led to dose reduction or discontinuation; all TRAEs were grade 1 in severity

Table 3. Patient-Level Antitumor Activity

Pt	Study	HER2 status	Prior HER2-targeted therapy	Zanidatamab dose	cBOR	PFS, months
6	FIH	IHC 3+/FISH+	Trastuzumab	20 mg/kg Q2W	PR	44.0+
7	FIH	IHC 3+/FISH+	None	20 mg/kg Q2W	PR	42.3+
3	FIH	IHC 3+/FISH+	None	20 mg/kg Q2W	PR	25.7
9	ZW25-102	IHC 3+/FISH+	None	1800 mg Q3W	PR	11.0
1	FIH	IHC 2+/FISH+	Trastuzumab + pertuzumab	20 mg/kg Q2W	SD	9.2
4	FIH	IHC 3+/FISH+	Trastuzumab	20 mg/kg Q2W	SD	9.1
2	FIH	IHC 3+/FISH+	Trastuzumab + pertuzumab	20 mg/kg Q2W	SD	3.8
5	FIH	IHC 3+/FISH+	Trastuzumab	20 mg/kg Q2W	SD	5.6
8	FIH	IHC 3+/FISH+	None	20 mg/kg Q2W	SD	3.7[a]
10	ZW25-204	IHC 3+/FISH+	Trastuzumab + pertuzumab; T-DM1	1600 mg Q2W + evorpacept	PR	18.4

+ indicates PFS was ongoing at data cutoff.
[a]Patient was lost to follow up.
cBOR, confirmed best overall response; FIH, first-in-human; FISH, fluorescence in situ hybridization; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; PFS, progression-free survival; PR, partial response; pt, patient; Q2W, every 2 weeks; Q3W, every 3 weeks; SD, stable disease; T-DM1, trastuzumab emtansine.

- Among patients who received zanidatamab monotherapy, the confirmed objective response rate (cORR) was 44% (95% CI: 14–79; 4 confirmed partial responses [PRs])
 - Among patients who were naïve to HER2-targeted therapies, the cORR was 75% (95% CI: 19–99; 3 PRs)
 - One PR (cORR, 20% [95% CI: 1–72]) and prolonged disease control were reported in patients with prior HER2-targeted therapies
- The patient who received zanidatamab + evorpacept (study ZW25-204) had a confirmed PR, a duration of response of 16.6 months, and a progression-free survival (PFS) of 18.4 months

Figure 3. Target Lesion Reduction in Patients With Advanced HER2-Positive SGC Treated With Zanidatamab Monotherapy

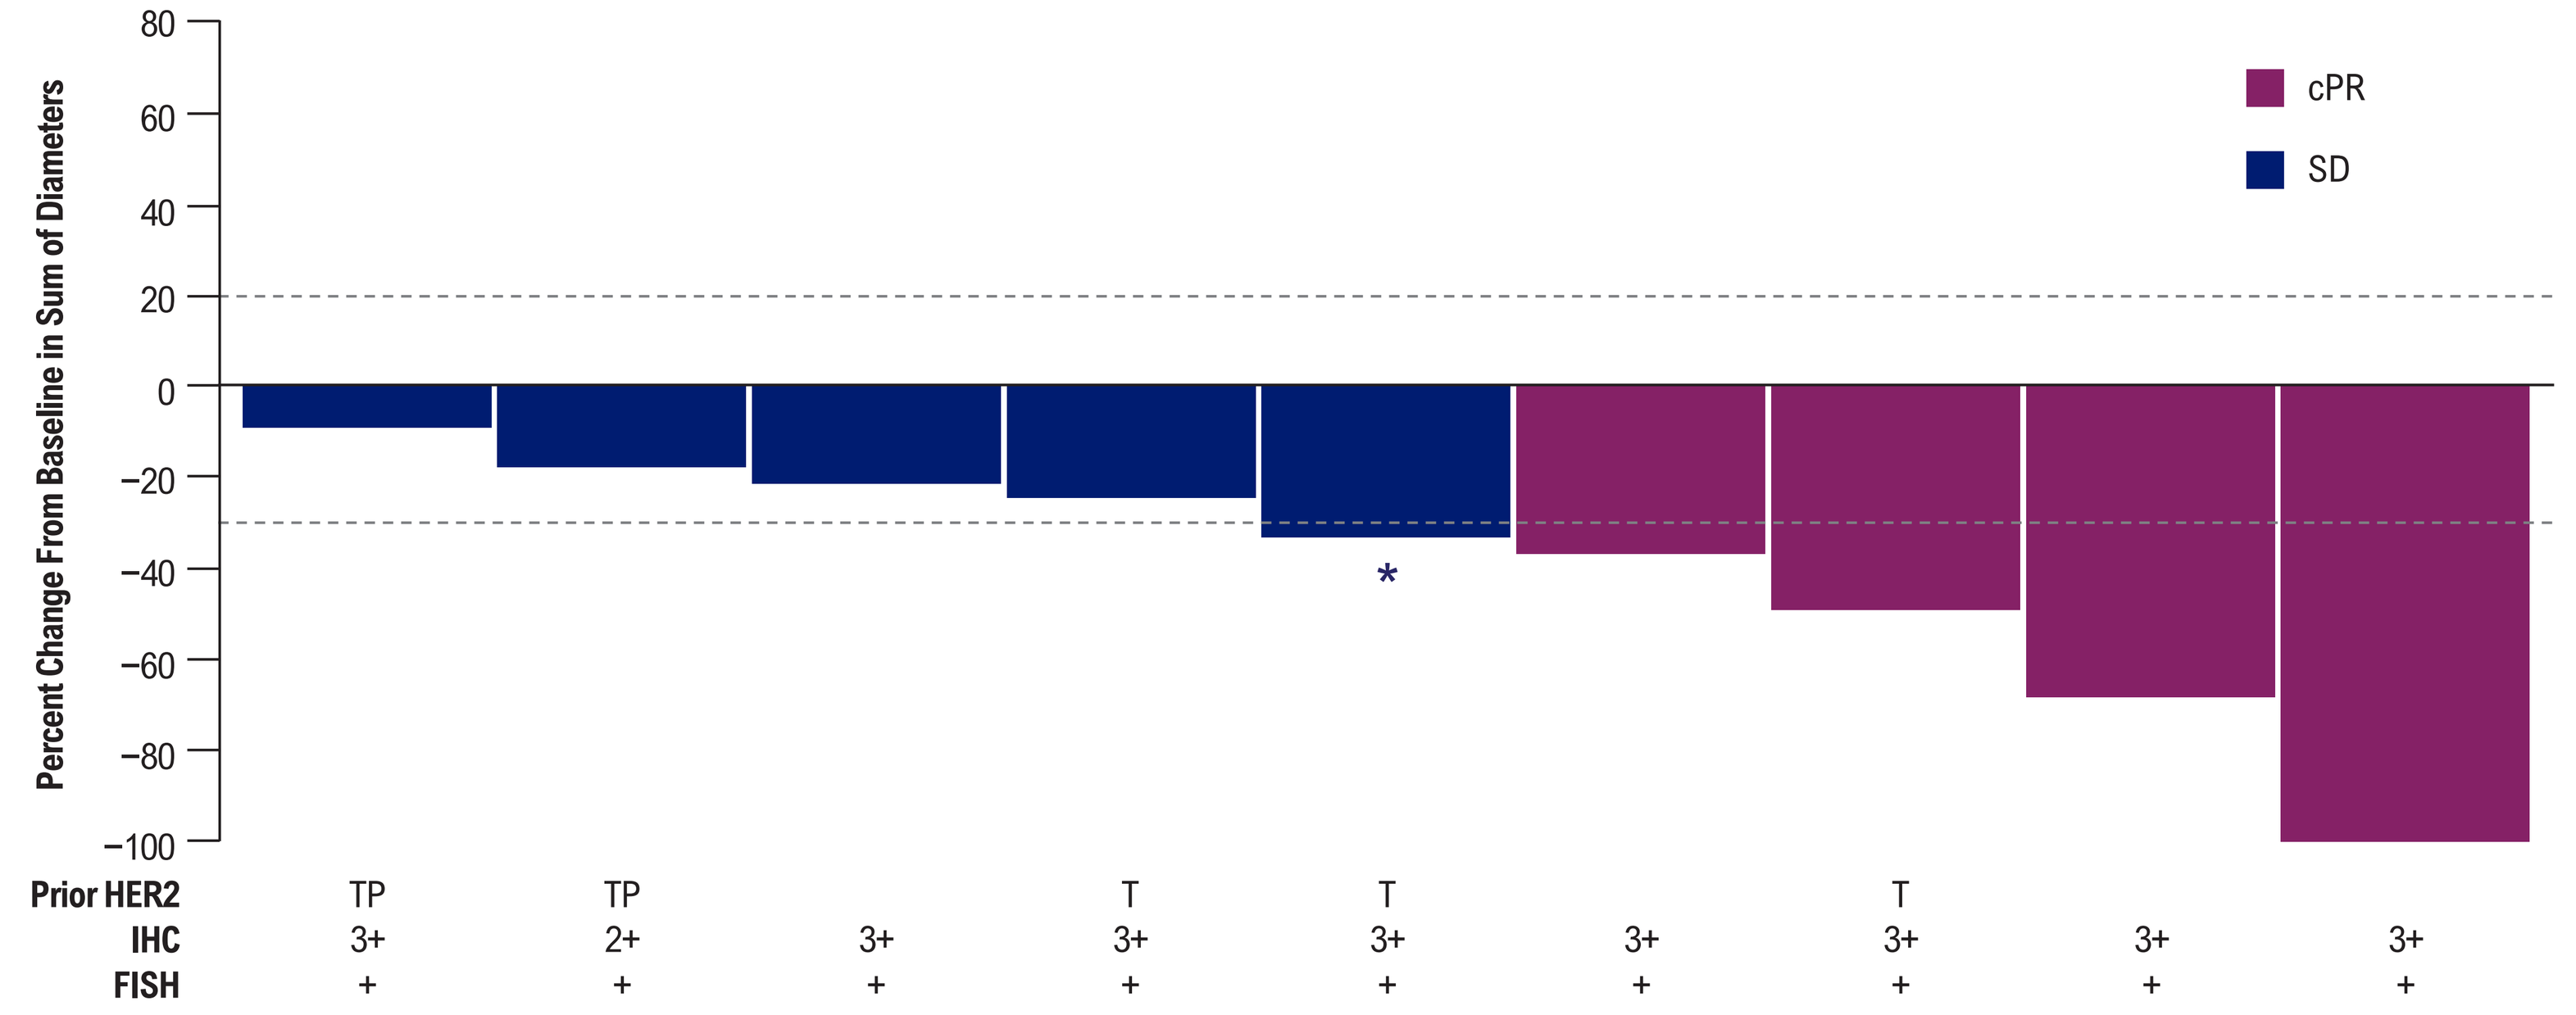

Percent Change From Baseline in Sum of Diameters
80
60
40
20
0
−20
−40
−60
−80
−100
cPR
SD
*
Prior HER2: TP, TP, , T, T, , T, ,
IHC: 3+, 2+, 3+, 3+, 3+, 3+, 3+, 3+, 3+
FISH: +, +, +, +, +, +, +, +, +

*Patient had a partial response, which was not confirmed. Dotted lines indicate thresholds for progressive disease (20% increase in sum of diameters of target lesions) and partial response (30% decrease in sum of diameters of target lesions) per RECIST v1.1.
cPR, confirmed partial response; FISH, fluorescence in situ hybridization; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; P, pertuzumab; RECIST v1.1, Response Evaluation Criteria in Solid Tumors version 1.1; SD, stable disease; SGC, salivary gland cancer; T, trastuzumab.

- All patients who received zanidatamab monotherapy had a reduction in tumor size (disease control rate, 100% [95% CI: 66–100])

Figure 4. Treatment Duration and Response in Patients With Advanced HER2-Positive SGC Treated With Zanidatamab Monotherapy

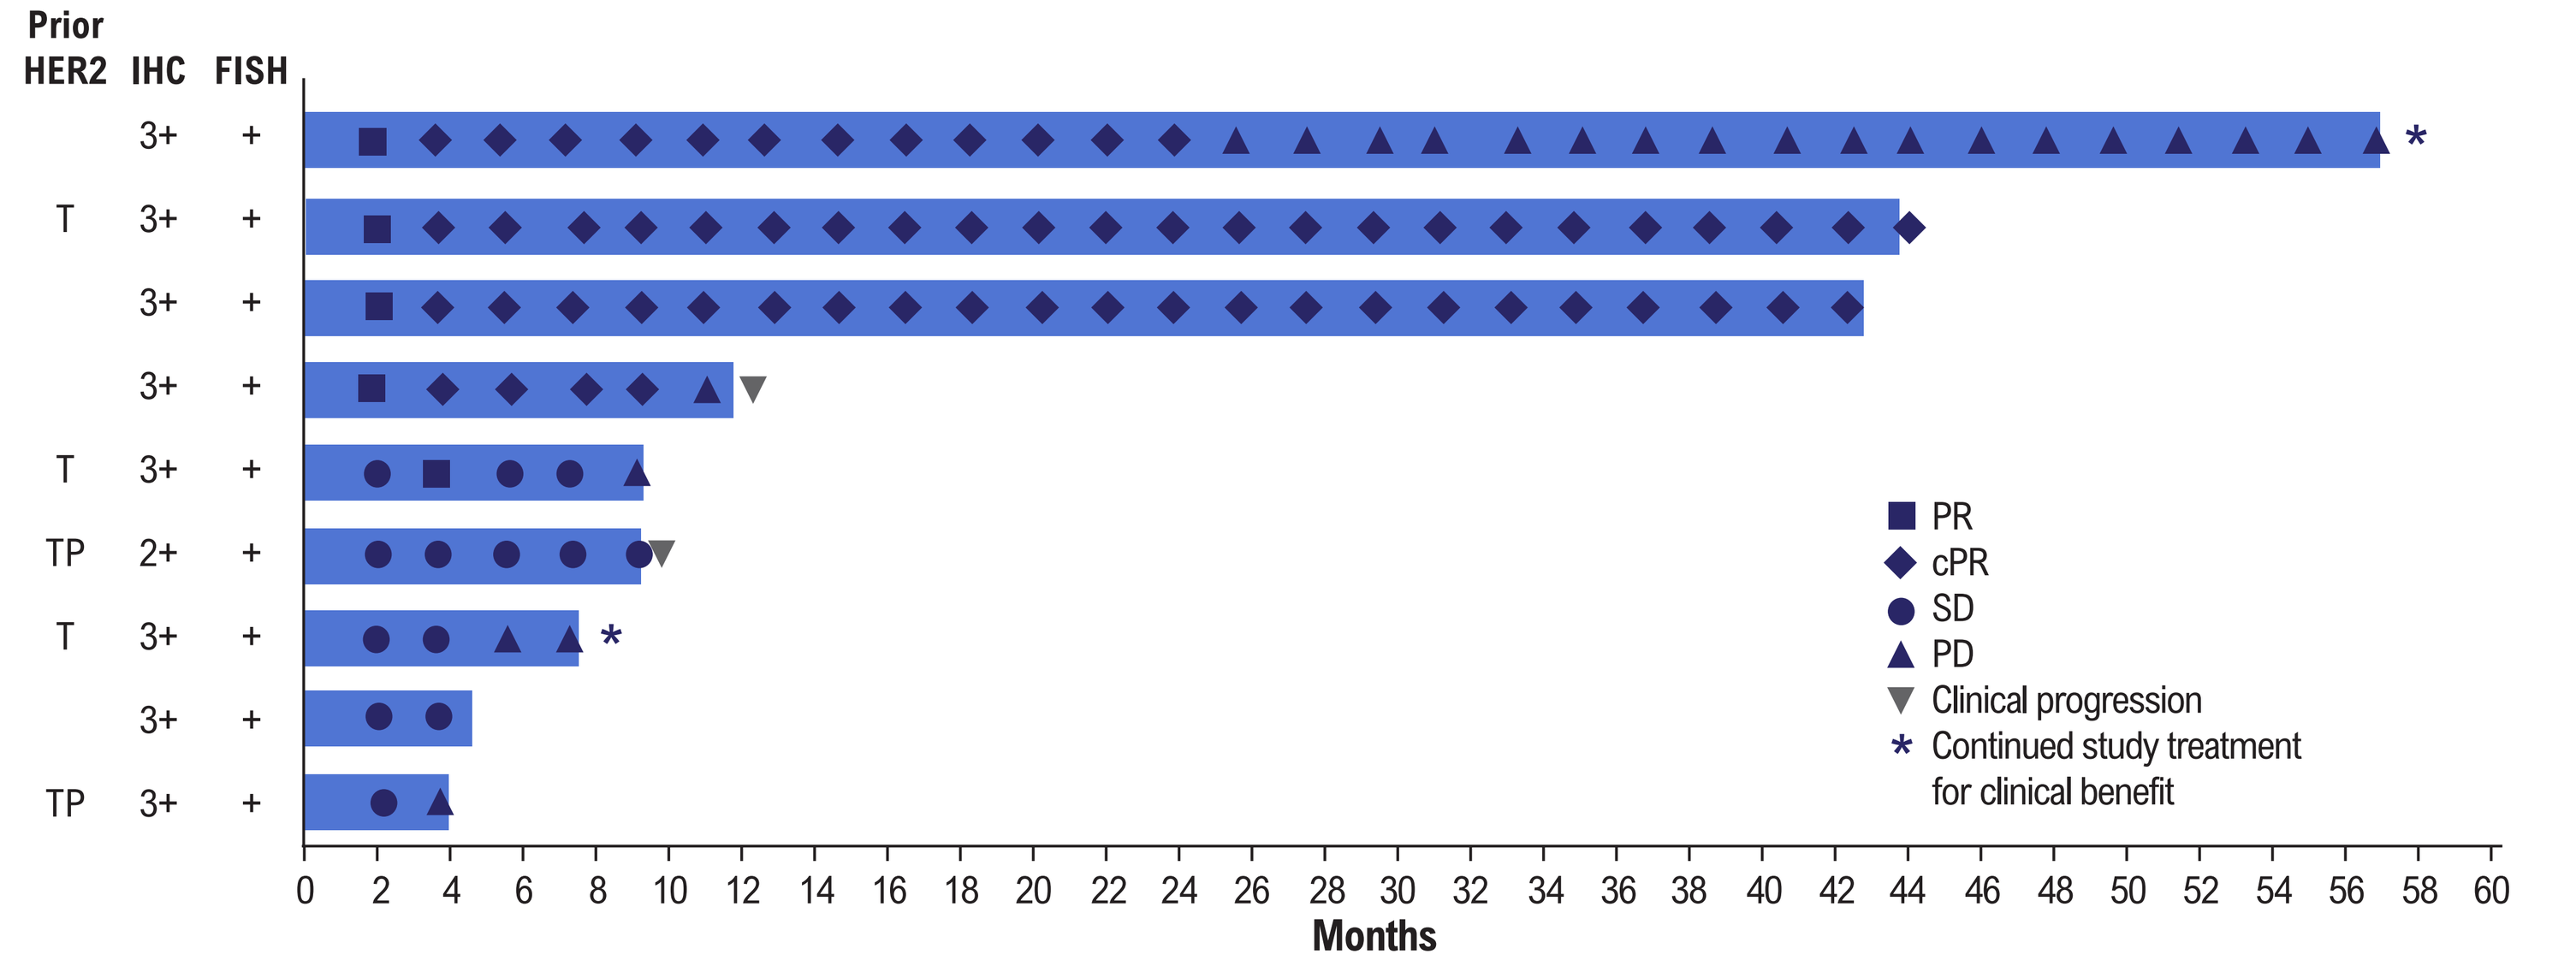

Prior HER2 | IHC | FISH
 | 3+ | +
T | 3+ | +
 | 3+ | +
 | 3+ | +
T | 3+ | +
TP | 2+ | +
T | 3+ | +
 | 3+ | +
TP | 3+ | +
PR
cPR
SD
PD
Clinical progression
Continued study treatment for clinical benefit
0 2 4 6 8 10 12 14 16 18 20 22 24 26 28 30 32 34 36 38 40 42 44 46 48 50 52 54 56 58 60
Months

cPR, confirmed PR; FISH, fluorescence in situ hybridization; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; P, pertuzumab; PD, progressive disease; PR, partial response; SD, stable disease; SGC, salivary gland cancer; T, trastuzumab.

- The 4 patients who achieved an objective response with zanidatamab monotherapy did so by their first assessment, with the response confirmed at the second assessment
 - The median duration of response was not estimable (NE)

Figure 5. Pooled Summary Kaplan-Meier Analysis of PFS in Patients With Advanced HER2-Positive SGC Treated With Zanidatamab Monotherapy

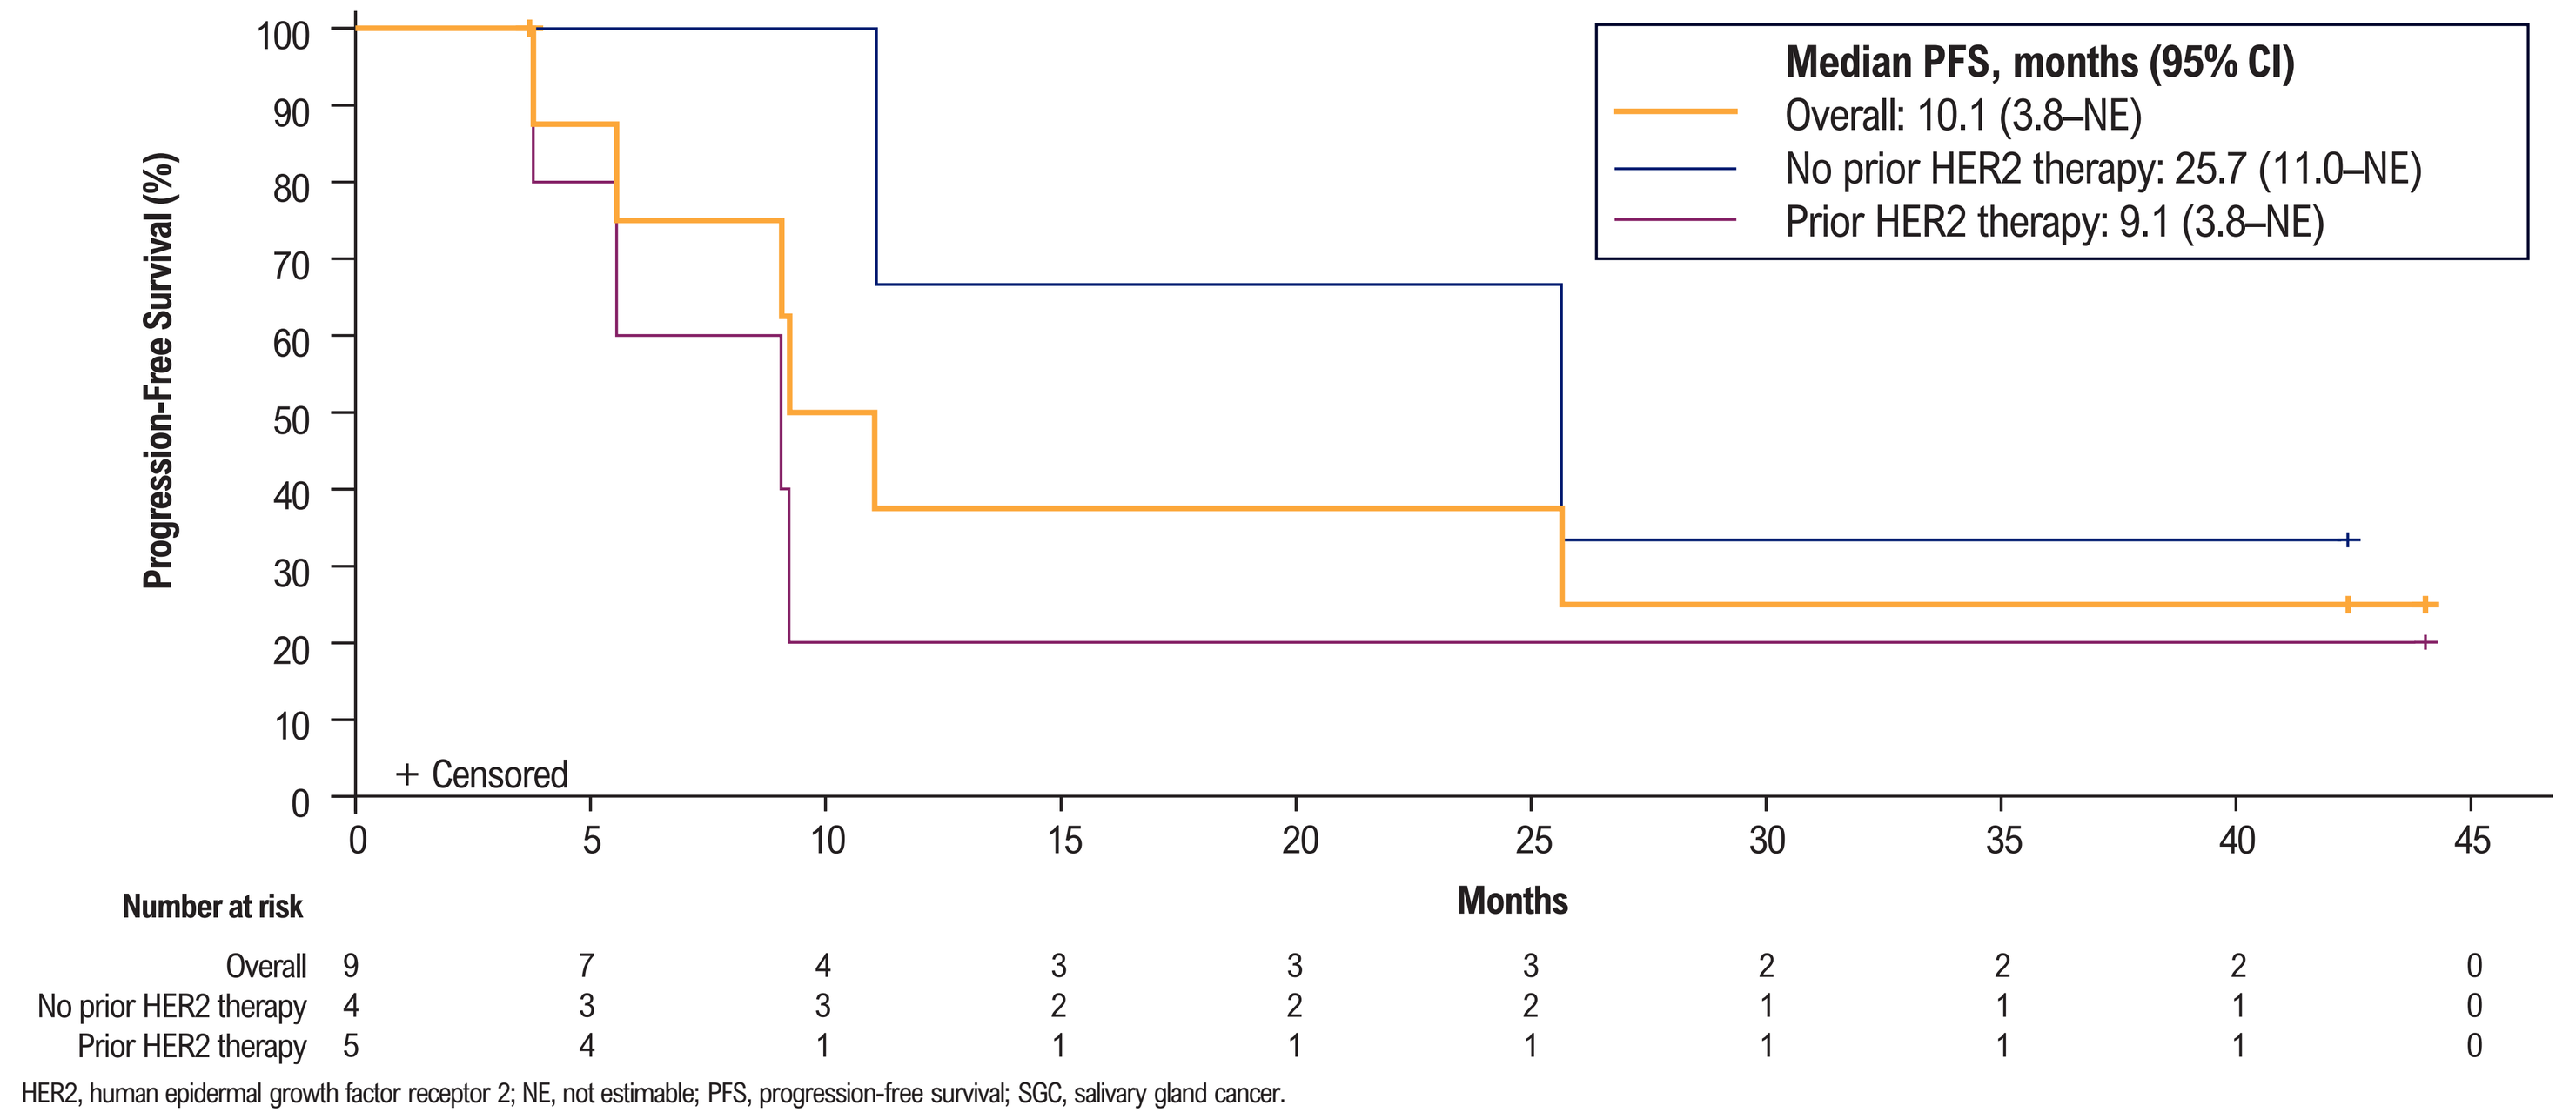

Progression-Free Survival (%)
100
90
80
70
60
50
40
30
20
10
0
Median PFS, months (95% CI)
Overall: 10.1 (3.8–NE)
No prior HER2 therapy: 25.7 (11.0–NE)
Prior HER2 therapy: 9.1 (3.8–NE)
+ Censored
0 5 10 15 20 25 30 35 40 45
Months
Number at risk
Overall 9 7 4 3 3 3 2 2 2 0
No prior HER2 therapy 4 3 3 2 2 2 1 1 1 0
Prior HER2 therapy 5 4 1 1 1 1 1 1 1 0

HER2, human epidermal growth factor receptor 2; NE, not estimable; PFS, progression-free survival; SGC, salivary gland cancer.

- The median PFS was 10.1 (95% CI: 3.8–NE) months

Conclusions

- Zanidatamab monotherapy was well tolerated, with only grade 1/2 TRAEs reported, and demonstrated encouraging preliminary antitumor activity in patients with heavily pretreated, HER2-positive advanced SGC
- All patients who received zanidatamab had a reduction in tumor size, regardless of prior HER2-targeted therapy
- Antitumor activity was shown with zanidatamab across dosing strategies (weight-based dosing, flat dosing, dosing every other week, and dosing every 3 weeks) and in combination with evorpacept, although sample sizes were limited
- These findings reinforce HER2 as an actionable target in SGC and support further evaluation of zanidatamab in HER2-positive SGC
 - The tumor-agnostic phase 2 DiscovHER PAN-206 study (NCT06695845) is currently evaluating zanidatamab in patients with previously treated HER2-overexpressing solid tumors, including SGC[7]

References: 1. Locati LD, et al. *Front Oncol*. 2023;13:1264287. **2.** Steuer CD, et al. *CA Cancer J Clin*. 2023;73:597-619. **3.** Egebjerg K, et al. *Front Oncol*. 2021;11:693394. **4.** Weisser NE, et al. *Nature Commun*. 2023;14(1):1394. **5.** Jazz Pharmaceuticals, Inc. Ziihera® (zanidatamab-hrii) for injection, for intravenous use. Updated November 2024. Accessed May 28, 2025. https://pp.jazzpharma.com/pi/ziihera.en.USPI.pdf. **6.** Meric-Bernstam F, et al. *Lancet Oncol*. 2022;23(12):1558-70. **7.** Jazz Pharmaceuticals. A phase 2 study of zanidatamab in patients with HER2-expressing tumors. ClinicalTrials.gov. Revised August 11, 2025. Accessed August 12, 2025. https://clinicaltrials.gov/study/NCT06695845

Support and Acknowledgments: Clinical research was funded by Jazz Pharmaceuticals. Under direction of the authors, Adam Schroer, MS, PhD, of Red Nucleus provided medical writing and editorial support, which was funded by Jazz Pharmaceuticals in accordance with Good Publication Practice (GPP 2022) guidelines (https://www.ismpp.org/gpp-2022). Jazz Pharmaceuticals also reviewed and edited the poster for scientific accuracy. The authors would like to thank all of the study investigators, study staff, nursing team, and patients for their participation in this research.

Disclosures: Funda Meric-Bernstam reports consulting or personal fees from AbbVie, Aduro BioTech, Alkermes, AstraZeneca, Chugai, DebioPharm, eFFECTOR Therapeutics, Genentech, IBM Watson, Infinity Pharmaceuticals, Jackson Laboratory, Kolon Life Science, Lengo Therapeutics, OrigiMed, PACT Pharma, Parexel International, Pfizer, Roche, Samsung Bioepis, Seagen, Tallac Therapeutics, Tyra Biosciences, Xencor, and Zymeworks; participating on advisory boards for Biovica, Black Diamond, Eisai, Immunomedics, Inflection Biosciences, Karyopharm Therapeutics, Loxo Oncology, Mersana Therapeutics, OnCusp Therapeutics, Puma Biotechnology, Seagen, Silverback Therapeutics, Spectrum Pharmaceuticals, and Zentalis; and receiving research support from Aileron Therapeutics, AstraZeneca, Bayer Healthcare Pharmaceutical, Calithera Biosciences, Curis, CytomX Therapeutics, Daiichi Sankyo, Debiopharm International, eFFECTOR Therapeutics, Genentech, Guardant Health, Klus Pharma, Novartis, Puma Biotechnology, Taiho Pharmaceutical, and Takeda Pharmaceutical.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

Corresponding author: Funda Meric-Bernstam
fmeric@mdanderson.org

Poster presented at the 2025 AACR-NCI-EORTC International Conference on Molecular Targets and Cancer Therapeutics; October 22–26, 2025; Boston, MA, USA.