307

Epidemiology and Diagnostic Landscape for Gastroesophageal Adenocarcinoma With a HER2 Focus

Andrew Ko[1], Wayne Su[2], Javier Sabater[3], Junji Lin[2*], Michelle Smith[4], Paranjoy Saharia[5], Ankit Rohilla[5], Geoffrey Ku[6]

[1]University of California San Francisco, San Francisco, CA, USA; [2]Jazz Pharmaceuticals, Philadelphia, PA, USA; [3]Jazz Pharmaceuticals, Oxford, UK; [4]Lumanity, London, UK; [5]Lumanity, Gurugram, India; [6]Memorial Sloan Kettering Cancer Center, New York, NY, USA

*Presenting author

Background

- Gastroesophageal adenocarcinoma (GEA) is a subset of gastroesophageal cancer (GEC) that comprises adenocarcinomas arising in the stomach (gastric), esophagus, and gastroesophageal junction (GEJ)[1]
- By anatomical subtype, an estimated 95% of gastric cancers (GCs) and 65% of esophageal/GEJ cancers are adenocarcinomas[2,3]
- There is limited evidence on the epidemiology of human epidermal growth factor receptor 2 (HER2)-positive tumors in patients with GEA and on laboratory diagnostic methods used to characterize HER2 positivity in patients with GEA

Objective

- To summarize existing evidence on the incidence, prevalence, and mortality rates associated with HER2-positive GEA, as well as the diagnostic methods used to detect HER2 positivity in patients with GEA

Methods

- Searches for a targeted literature review were conducted using Embase.com for literature published between January 1, 2014, and July 1, 2024, covering Embase and MEDLINE, without restrictions on country or language
- Titles/abstracts and full-text papers were screened according to the predefined inclusion criteria. Screening was performed by a single reviewer, with a 20% random quality check conducted by a second senior reviewer on completion of the screening process

Table 1. Inclusion Criteria

Category	Inclusion Criteria
Population	Adults (≥18 years) diagnosed with GEA, including gastric, esophageal, and GEJ subtypes, with HER2-positive tumors
Outcomes	• Epidemiology (incidence, prevalence, and mortality) of HER2-positive status in GEA population • Diagnostic concordance between IHC and any other methods used to detect HER2 positivity • Overlap between HER2 positivity and PD-L1 expression (ie, as indicated by the proportion of patients with HER2-positive tumors who also present with a CPS of ≥1)
Study design	• Cohort, case-control, and cross-sectional studies • RCTs, nRCTs, and single-arm trials[a]
Time frame	January 1, 2014, to July 1, 2024

[a]RCTs, nRCTs, single-arm trials, and case-control studies were included for the publications assessing patients with HER2 overexpression and expression with amplification in GEA.
CPS, combined positive score; GEA, gastroesophageal adenocarcinoma; GEJ, gastroesophageal junction; HER2, human epidermal growth factor receptor 2; IHC, immunohistochemistry; nRCT, nonrandomized controlled trial; PD-L1, programmed death-ligand 1; RCT, randomized controlled trial.

- Acknowledging the paucity of epidemiological evidence from database searches on HER2 positivity rates in the GEA population, Global Cancer Observatory (GLOBOCAN) 2022 data were used to inform the incidence, prevalence, and mortality of all-comers GC and esophageal cancer, irrespective of HER2-positive status, for major geographies, namely Canada, Japan, the UK, the US, and the European Union 4 (EU4; ie, France, Germany, Italy, Spain)
- Furthermore, weighted average prevalence, incidence, and mortality rates for GEA were estimated for the US by using combined data from National Cancer Institute (NCI) Surveillance, Epidemiology, and End Results Program (SEER)[4,5]

Results

Figure 1. Study Selection PRISMA Flowchart

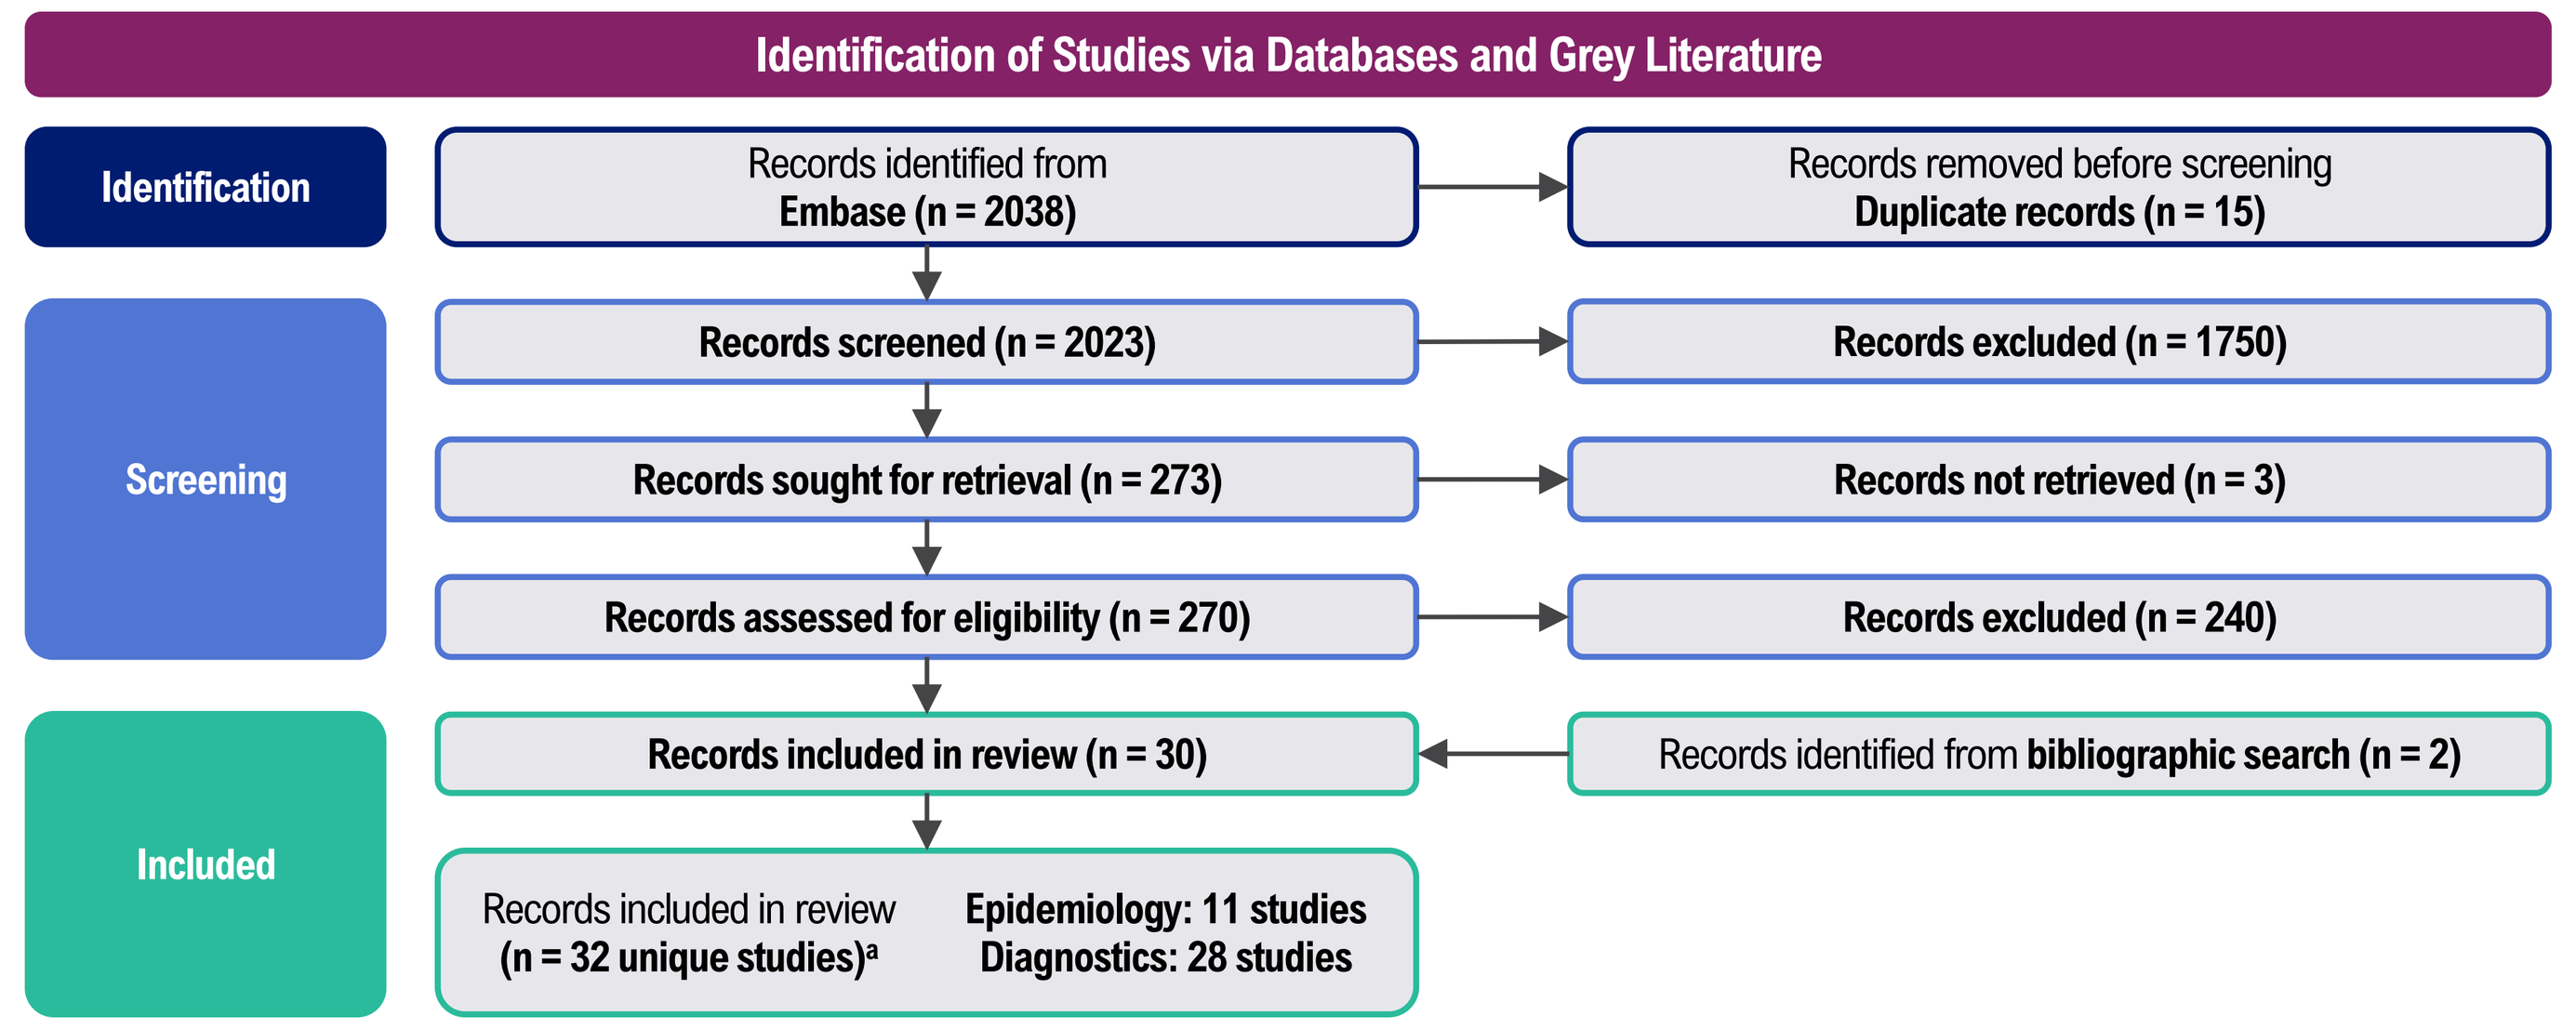

Identification of Studies via Databases and Grey Literature
Identification
Records identified from Embase (n = 2038)
Records removed before screening Duplicate records (n = 15)
Screening
Records screened (n = 2023)
Records excluded (n = 1750)
Records sought for retrieval (n = 273)
Records not retrieved (n = 3)
Records assessed for eligibility (n = 270)
Records excluded (n = 240)
Included
Records included in review (n = 30)
Records identified from bibliographic search (n = 2)
Records included in review (n = 32 unique studies)[a]
Epidemiology: 11 studies
Diagnostics: 28 studies

[a]In addition, GLOBOCAN 2022 data were retrieved to extract epidemiological data in the "all-comers" (ie, irrespective of HER2-positive status) GC/esophageal cancer population, and 2025 NCI SEER data were retrieved to estimate incidence rates specific to the US.
GC, gastric cancer; GLOBOCAN, Global Cancer Observatory; HER2, human epidermal growth factor receptor 2; NCI, National Cancer Institute; PRISMA, Preferred Reporting Items for Systematic Reviews and Meta-Analyses; SEER, Surveillance, Epidemiology, and End Results Program.

- Of the 2038 records retrieved from Embase.com and the bibliographic searches, 32 relevant studies were identified as containing epidemiological and/or diagnostic evidence. These included 11 studies with epidemiological evidence of HER2-positive status in a GEA population and 28 studies with diagnostic insights; 7 studies reported both epidemiological and diagnostic outcomes for patients with HER2-positive GEA

Table 2. Summary of Studies Reporting the Prevalence of HER2-Positive Status

Study Reference	Study Design; Data Sources	Study Period	Sample Size	Prevalence (%)	Country Coverage
GEA					
Lau-Min 2020[6]	Retrospective observational; Flatiron Health Oncology Database	2011–2019	6479	15.0	US
GC					
Cordero-García 2019[7]	Retrospective observational; hospital records	2010–2012 2012–2015	112	8.1 4.0	Costa Rica
Fleitas 2023[8]	Prospective observational; NR	2019–2023	148	Overall: 9.0 Latin America: 6.0 Europe: 14.0	Latin America and Europe
Gurbuz 2022[9]	Retrospective observational; hospital records	2017–2021	552	18.1	Turkey
Chong 2016[10]	Cross-sectional; hospital records	NR	103	13.6	Brunei Darussalam
Gupta 2016[11]	Prospective observational; hospital records	2013–2015	110	24.5	India
Kattan 2022[12]	Cross-sectional; hospital records	2011–2014	157	21.0	Lebanon
Li 2023[13]	Prospective observational; hospital records	2016–2020	238	47.9	China
GC/GEJC					
Nakayama 2023[14]	Retrospective observational; hospital records	2011–2018	734	23.2	Japan
Rajadurai 2018[15]	Retrospective and prospective observational; hospital records	2013–2014	228	24.6	Malaysia
Seo 2017[16]	Retrospective observational; NR	2011–2013	1687	10.8	Republic of Korea

GC, gastric cancer; GEA, gastroesophageal adenocarcinoma; GEJC, gastroesophageal junction cancer; HER2, human epidermal growth factor receptor 2; NR, not reported.

- Limited prevalence data were available for patients with GEA and a HER2-positive diagnosis; only 1 study reported HER2-positive prevalence within the overall GEA population, while the remaining 10 studies reported HER2-positive prevalence in patients with GC or GEJ cancer (GEJC)
- No incidence or mortality data were identified for HER2-positive patients within the overall GEA population
- A multicountry study including data from 2019–2023 from Europe (Spain, the Netherlands and Portugal) and Latin America (Argentina, Mexico, Chile, and Paraguay) reported an overall HER2-positive status prevalence rate of 9% in patients with GC, with the proportion being higher in Europe (14%) compared with Latin America (6%)[8]
- A similar HER2-positive prevalence of 4%–8% was observed in another study conducted in Latin America (Costa Rica)[7]
- In the US, a study of 6479 patients with GEA reported a HER2-positive prevalence of 15%[6]
- In Asia, prevalence of HER2-positive status for patients with GC ranged from 13.6% in Brunei Darussalam[10] to 47.9% in a study in China[13]

Figure 2. Age-Standardized Incidence of (A) GC and (B) Esophageal Cancer by Country

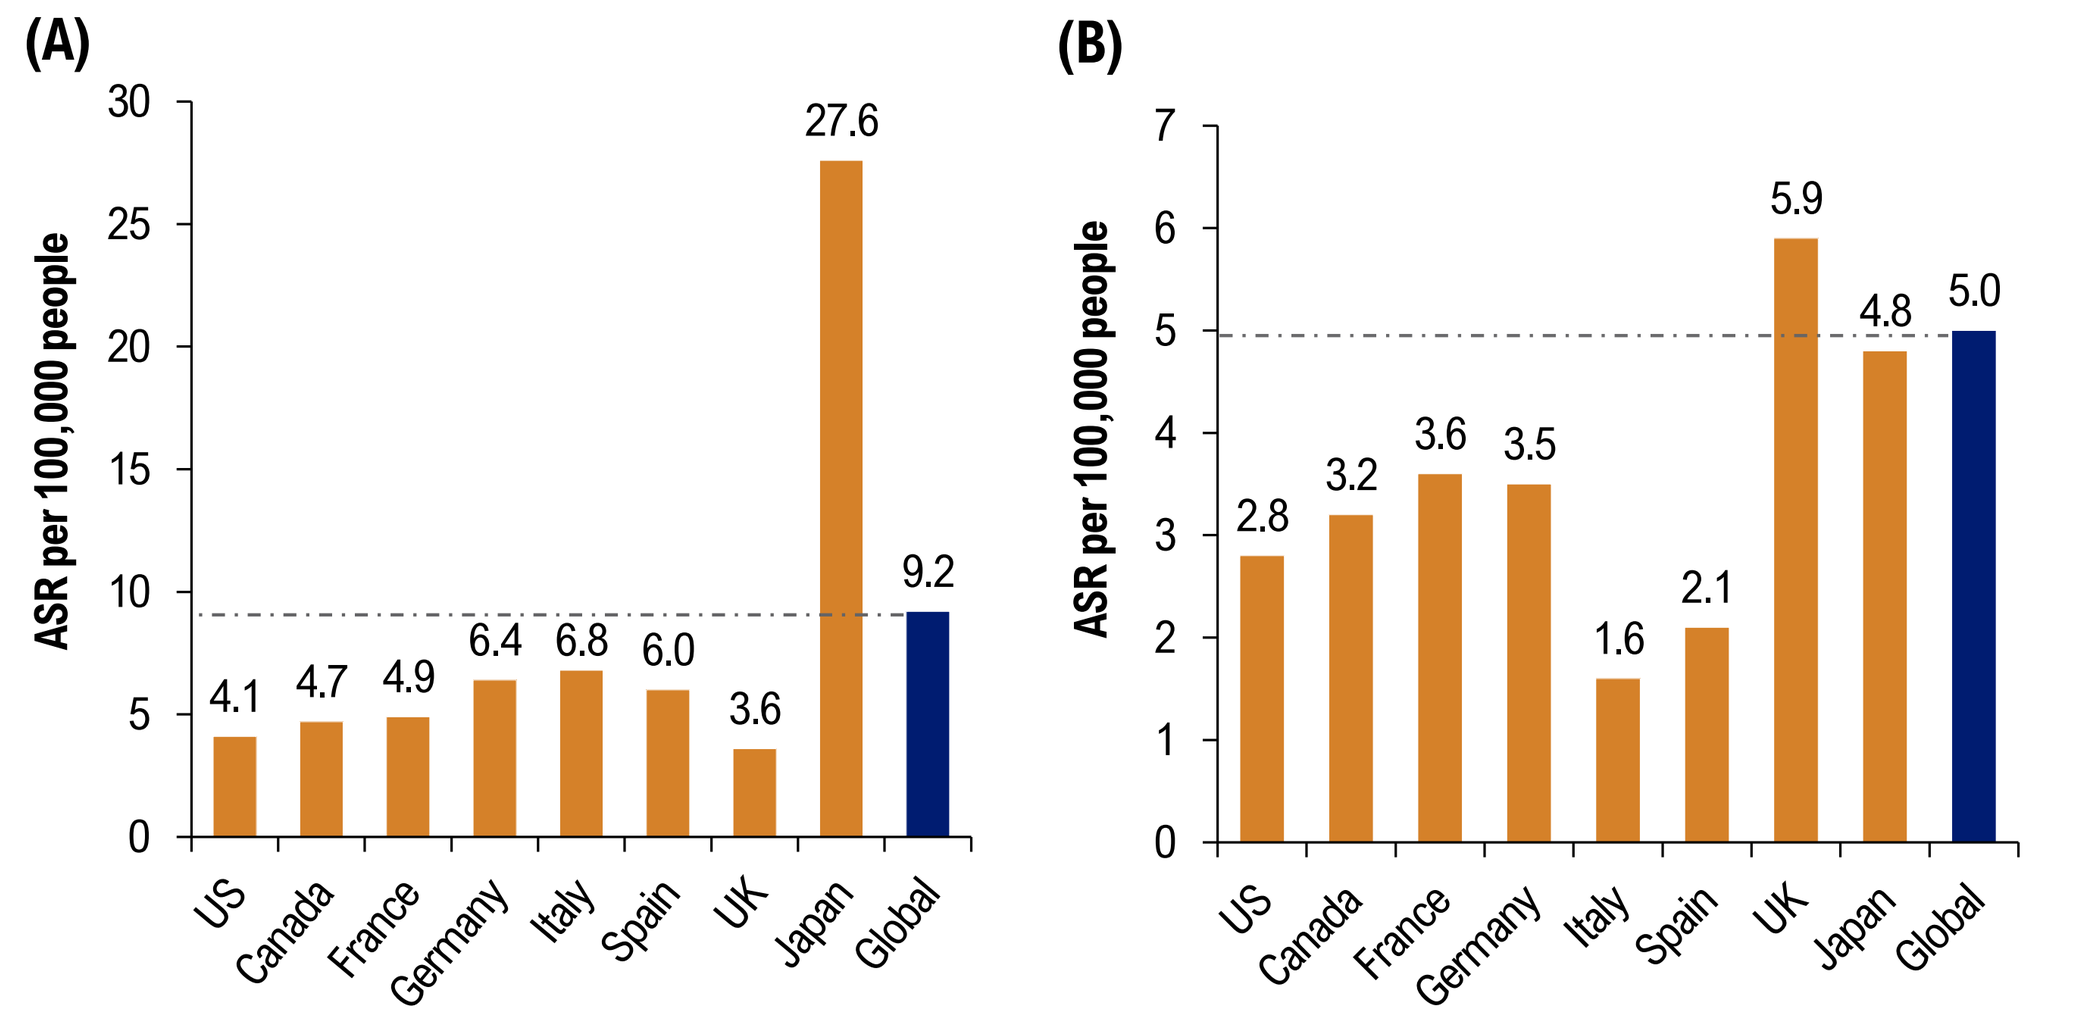

(A)
ASR per 100,000 people
0
5
10
15
20
25
30
4.1
4.7
4.9
6.4
6.8
6.0
3.6
27.6
9.2
US
Canada
France
Germany
Italy
Spain
UK
Japan
Global
(B)
ASR per 100,000 people
0
1
2
3
4
5
6
7
2.8
3.2
3.6
3.5
1.6
2.1
5.9
4.8
5.0
US
Canada
France
Germany
Italy
Spain
UK
Japan
Global

Source: GLOBOCAN.[17]
The dashed line is indicative of global data in each graph.
ASR, age-standardized rate; GC, gastric cancer; GLOBOCAN, Global Cancer Observatory.

In the absence of data specific to GEA, epidemiological data for all-comers GC or esophageal cancer (irrespective of HER2-positive status) in the general population were retrieved from GLOBOCAN data published in 2022.[17] With 95% of GC and 65% of esophageal/GEJ cancers considered to be adenocarcinomas,[2,3] the epidemiological data from GLOBOCAN are indicative of the GEA epidemiology

- The global crude incidence rate for GC was 12.3 cases per 100,000 people, with an age-standardized rate (ASR) of 9.2 cases per 100,000 people
 - The age-standardized incidence rate for GC was considerably higher in Japan (27.6 per 100,000 people) compared with the US, Canada, the UK, and the EU4 (3.6–6.8 per 100,000 people)
- For esophageal cancer, the crude incidence rate was 6.5 cases per 100,000 people (ASR: 5 cases per 100,000 people)
- Incidence rates were higher in males than females for GC (crude: 15.8 vs 8.7 per 100,000 people; ASR: 12.8 vs 6.0 per 100,000 people) and esophageal cancer (crude: 9.2 vs 3.7 per 100,000 people; ASR: 7.6 vs 2.6 per 100,000 people; data not shown)

Figure 3. Five-Year Prevalence Rates of (A) GC and (B) Esophageal Cancer by Country

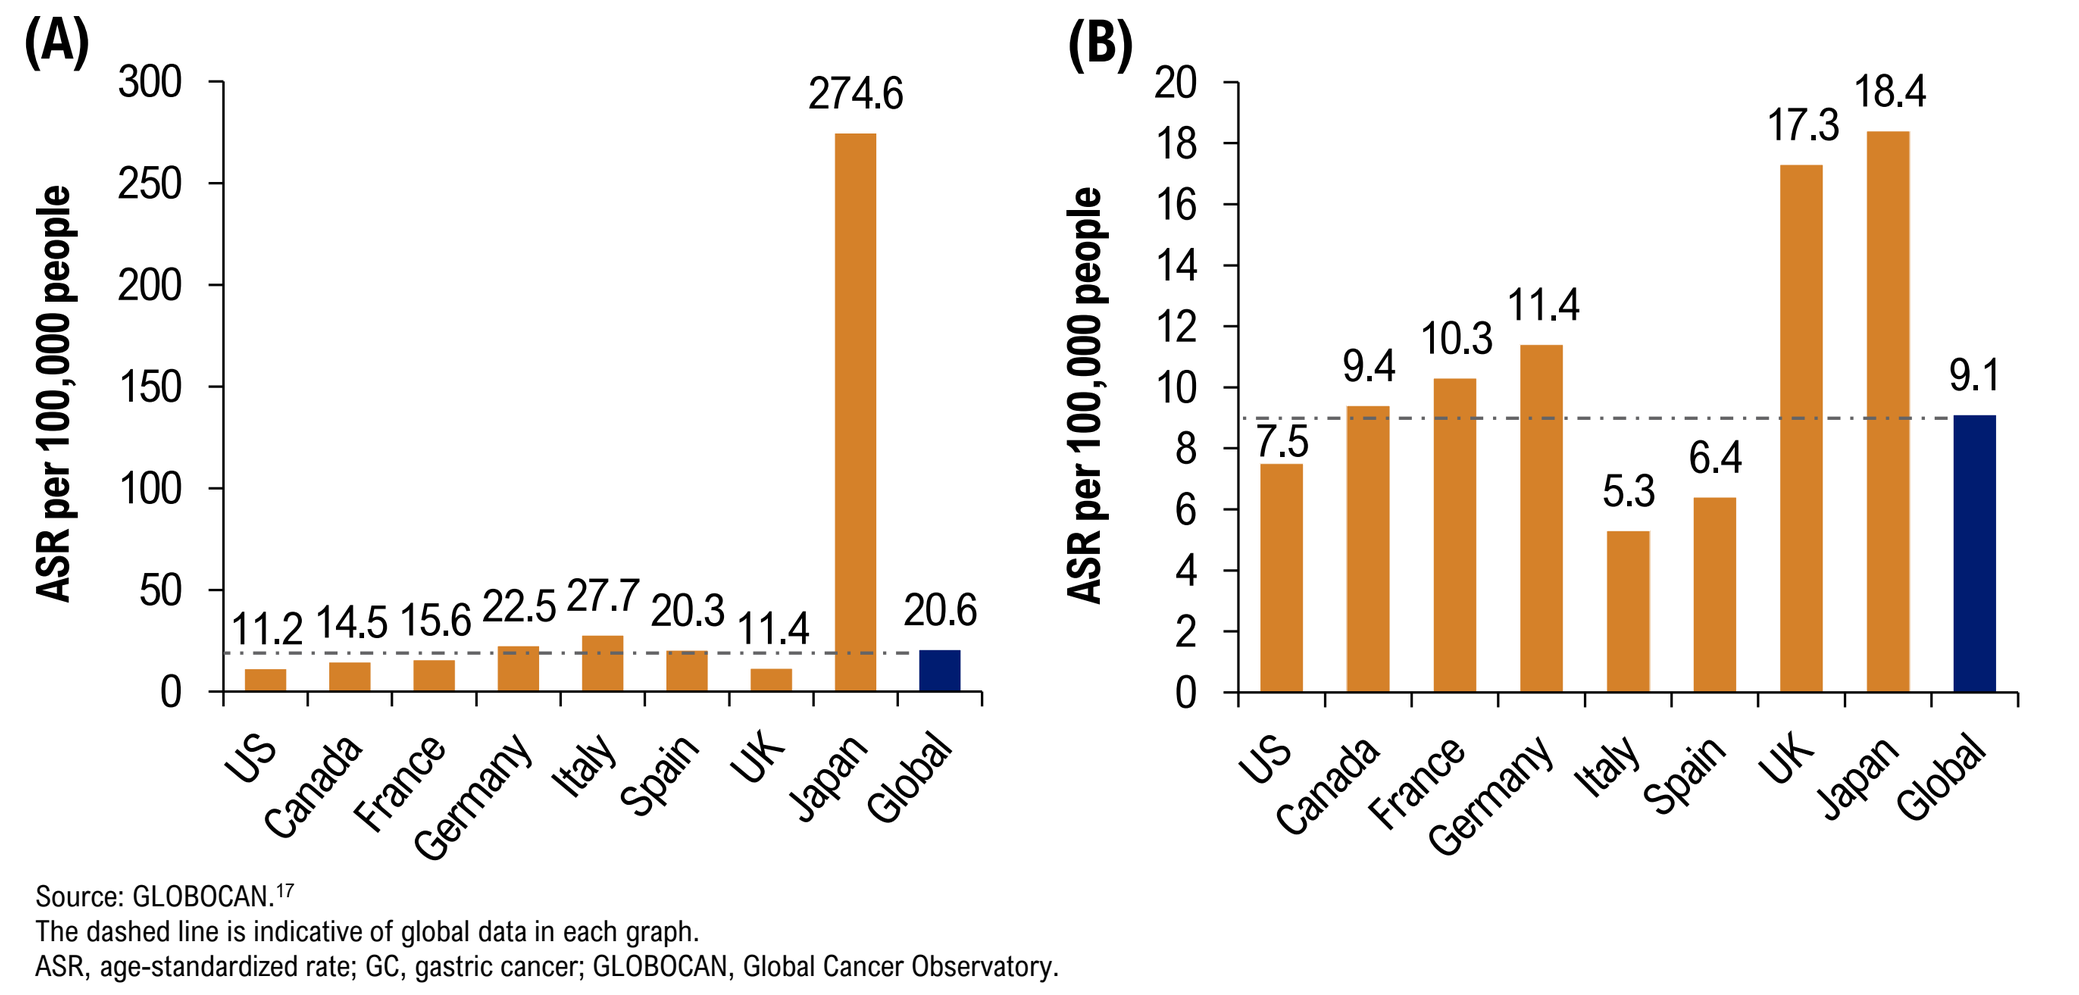

(A)
ASR per 100,000 people
0
50
100
150
200
250
300
11.2
14.5
15.6
22.5
27.7
20.3
11.4
274.6
20.6
US
Canada
France
Germany
Italy
Spain
UK
Japan
Global
(B)
ASR per 100,000 people
0
2
4
6
8
10
12
14
16
18
20
7.5
9.4
10.3
11.4
5.3
6.4
17.3
18.4
9.1
US
Canada
France
Germany
Italy
Spain
UK
Japan
Global

Source: GLOBOCAN.[17]
The dashed line is indicative of global data in each graph.
ASR, age-standardized rate; GC, gastric cancer; GLOBOCAN, Global Cancer Observatory.

- Globally, the 5-year prevalence rates of GC were 20.6 per 100,000. Rates were considerably higher in Japan (274.6 per 100,000 people) compared with the US, Canada, the UK, and the EU4 (11.2–27.7 per 100,000 people)
- The 5-year prevalence rate of esophageal cancer globally was 9.1 per 100,000 people. Rates were higher in Japan (18.4 per 100,000 people) and the UK (17.3 per 100,000 people) compared with the other geographies
- The 5-year prevalence rates were considerably higher among males compared with females for GC (26.3 vs 14.9 cases per 100,000 people) as well as esophageal cancer (12.8 vs 5.4 cases per 100,000 people; data not shown)

Figure 4. Age-Standardized Mortality Rates of (A) GC and (B) Esophageal Cancer by Country

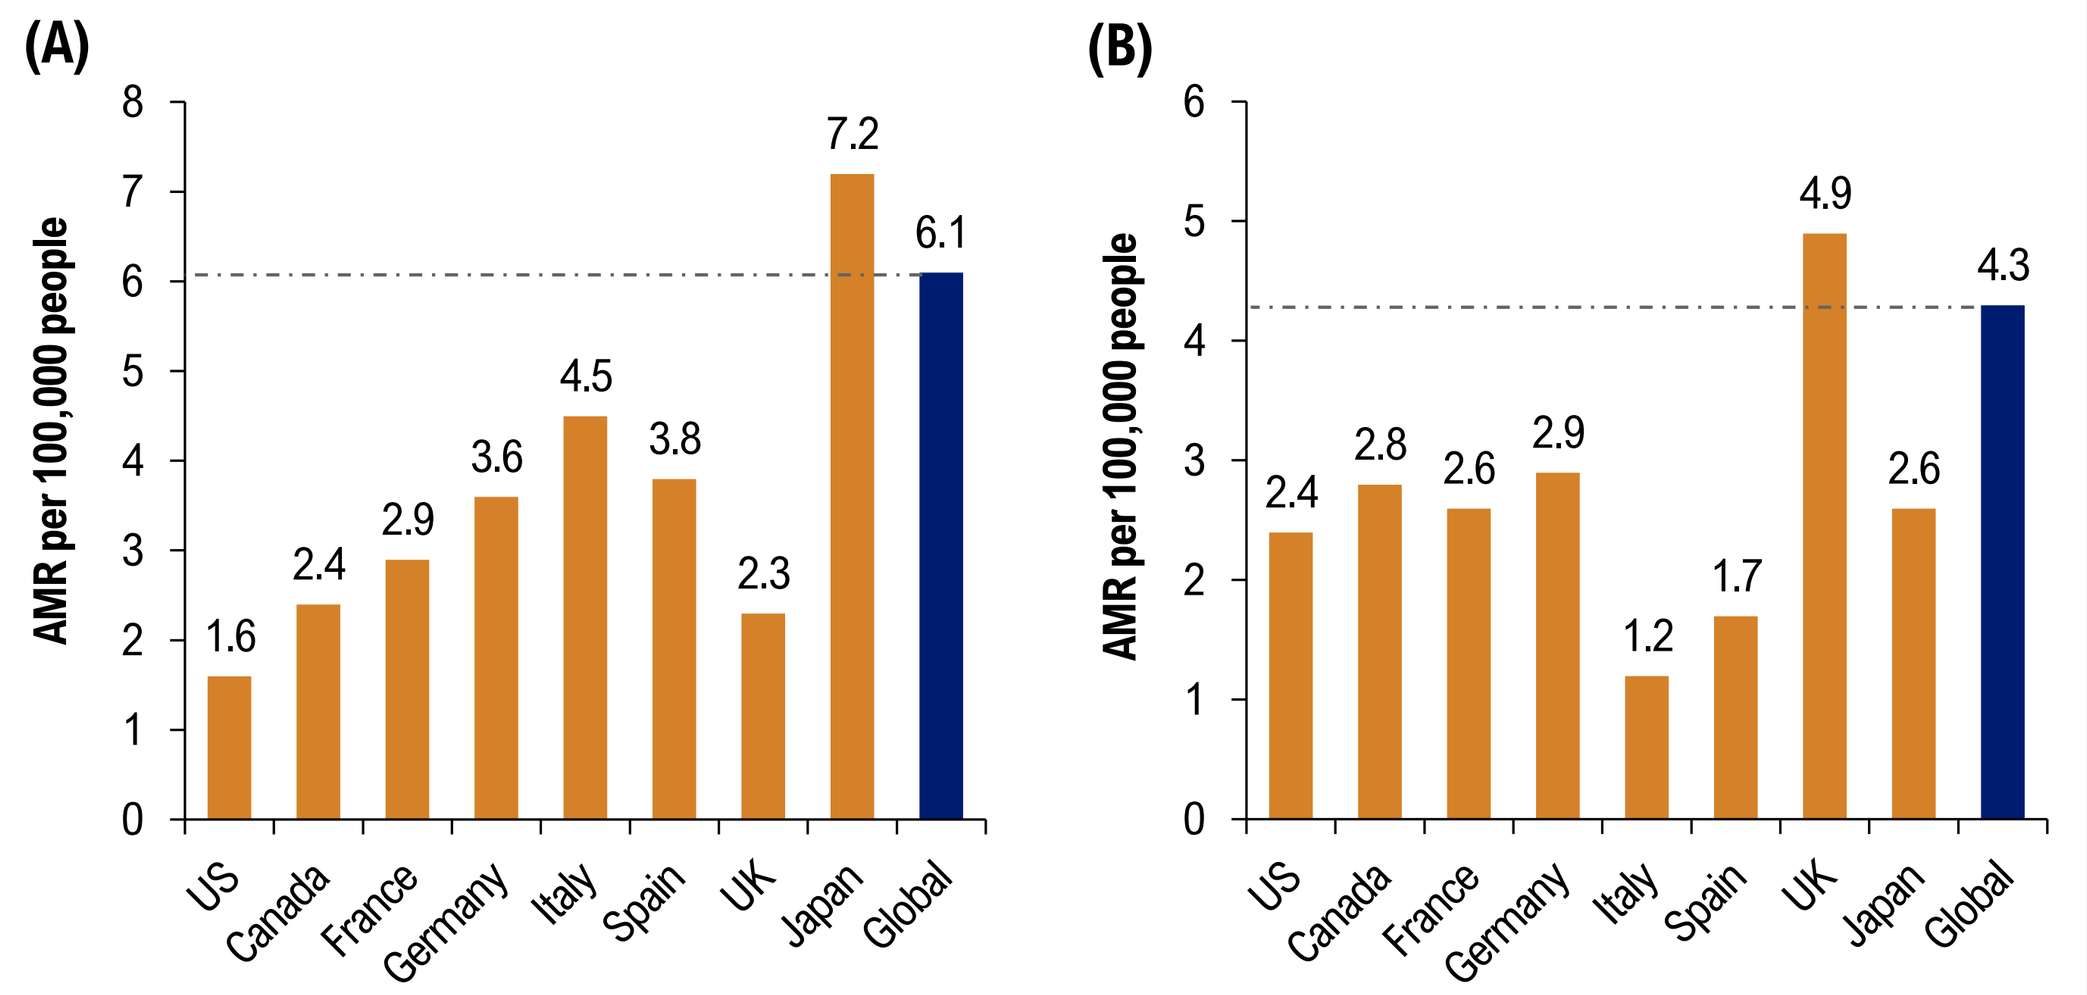

(A)
AMR per 100,000 people
0
1
2
3
4
5
6
7
8
1.6
2.4
2.9
3.6
4.5
3.8
2.3
7.2
6.1
US
Canada
France
Germany
Italy
Spain
UK
Japan
Global
(B)
AMR per 100,000 people
0
1
2
3
4
5
6
2.4
2.8
2.6
2.9
1.2
1.7
4.9
2.6
4.3
US
Canada
France
Germany
Italy
Spain
UK
Japan
Global

Source: GLOBOCAN.[17]
The dashed line is indicative of global data in each graph.
AMR, age-standardized mortality rate; GC, gastric cancer; GLOBOCAN, Global Cancer Observatory.

- The global age-standardized mortality rate was 6.1 cases per 100,000 people for patients with GC and 4.3 cases per 100,000 people for patients with esophageal cancer

Figure 5. Incidence of HER2-Positive Advanced/Metastatic GEA in the US

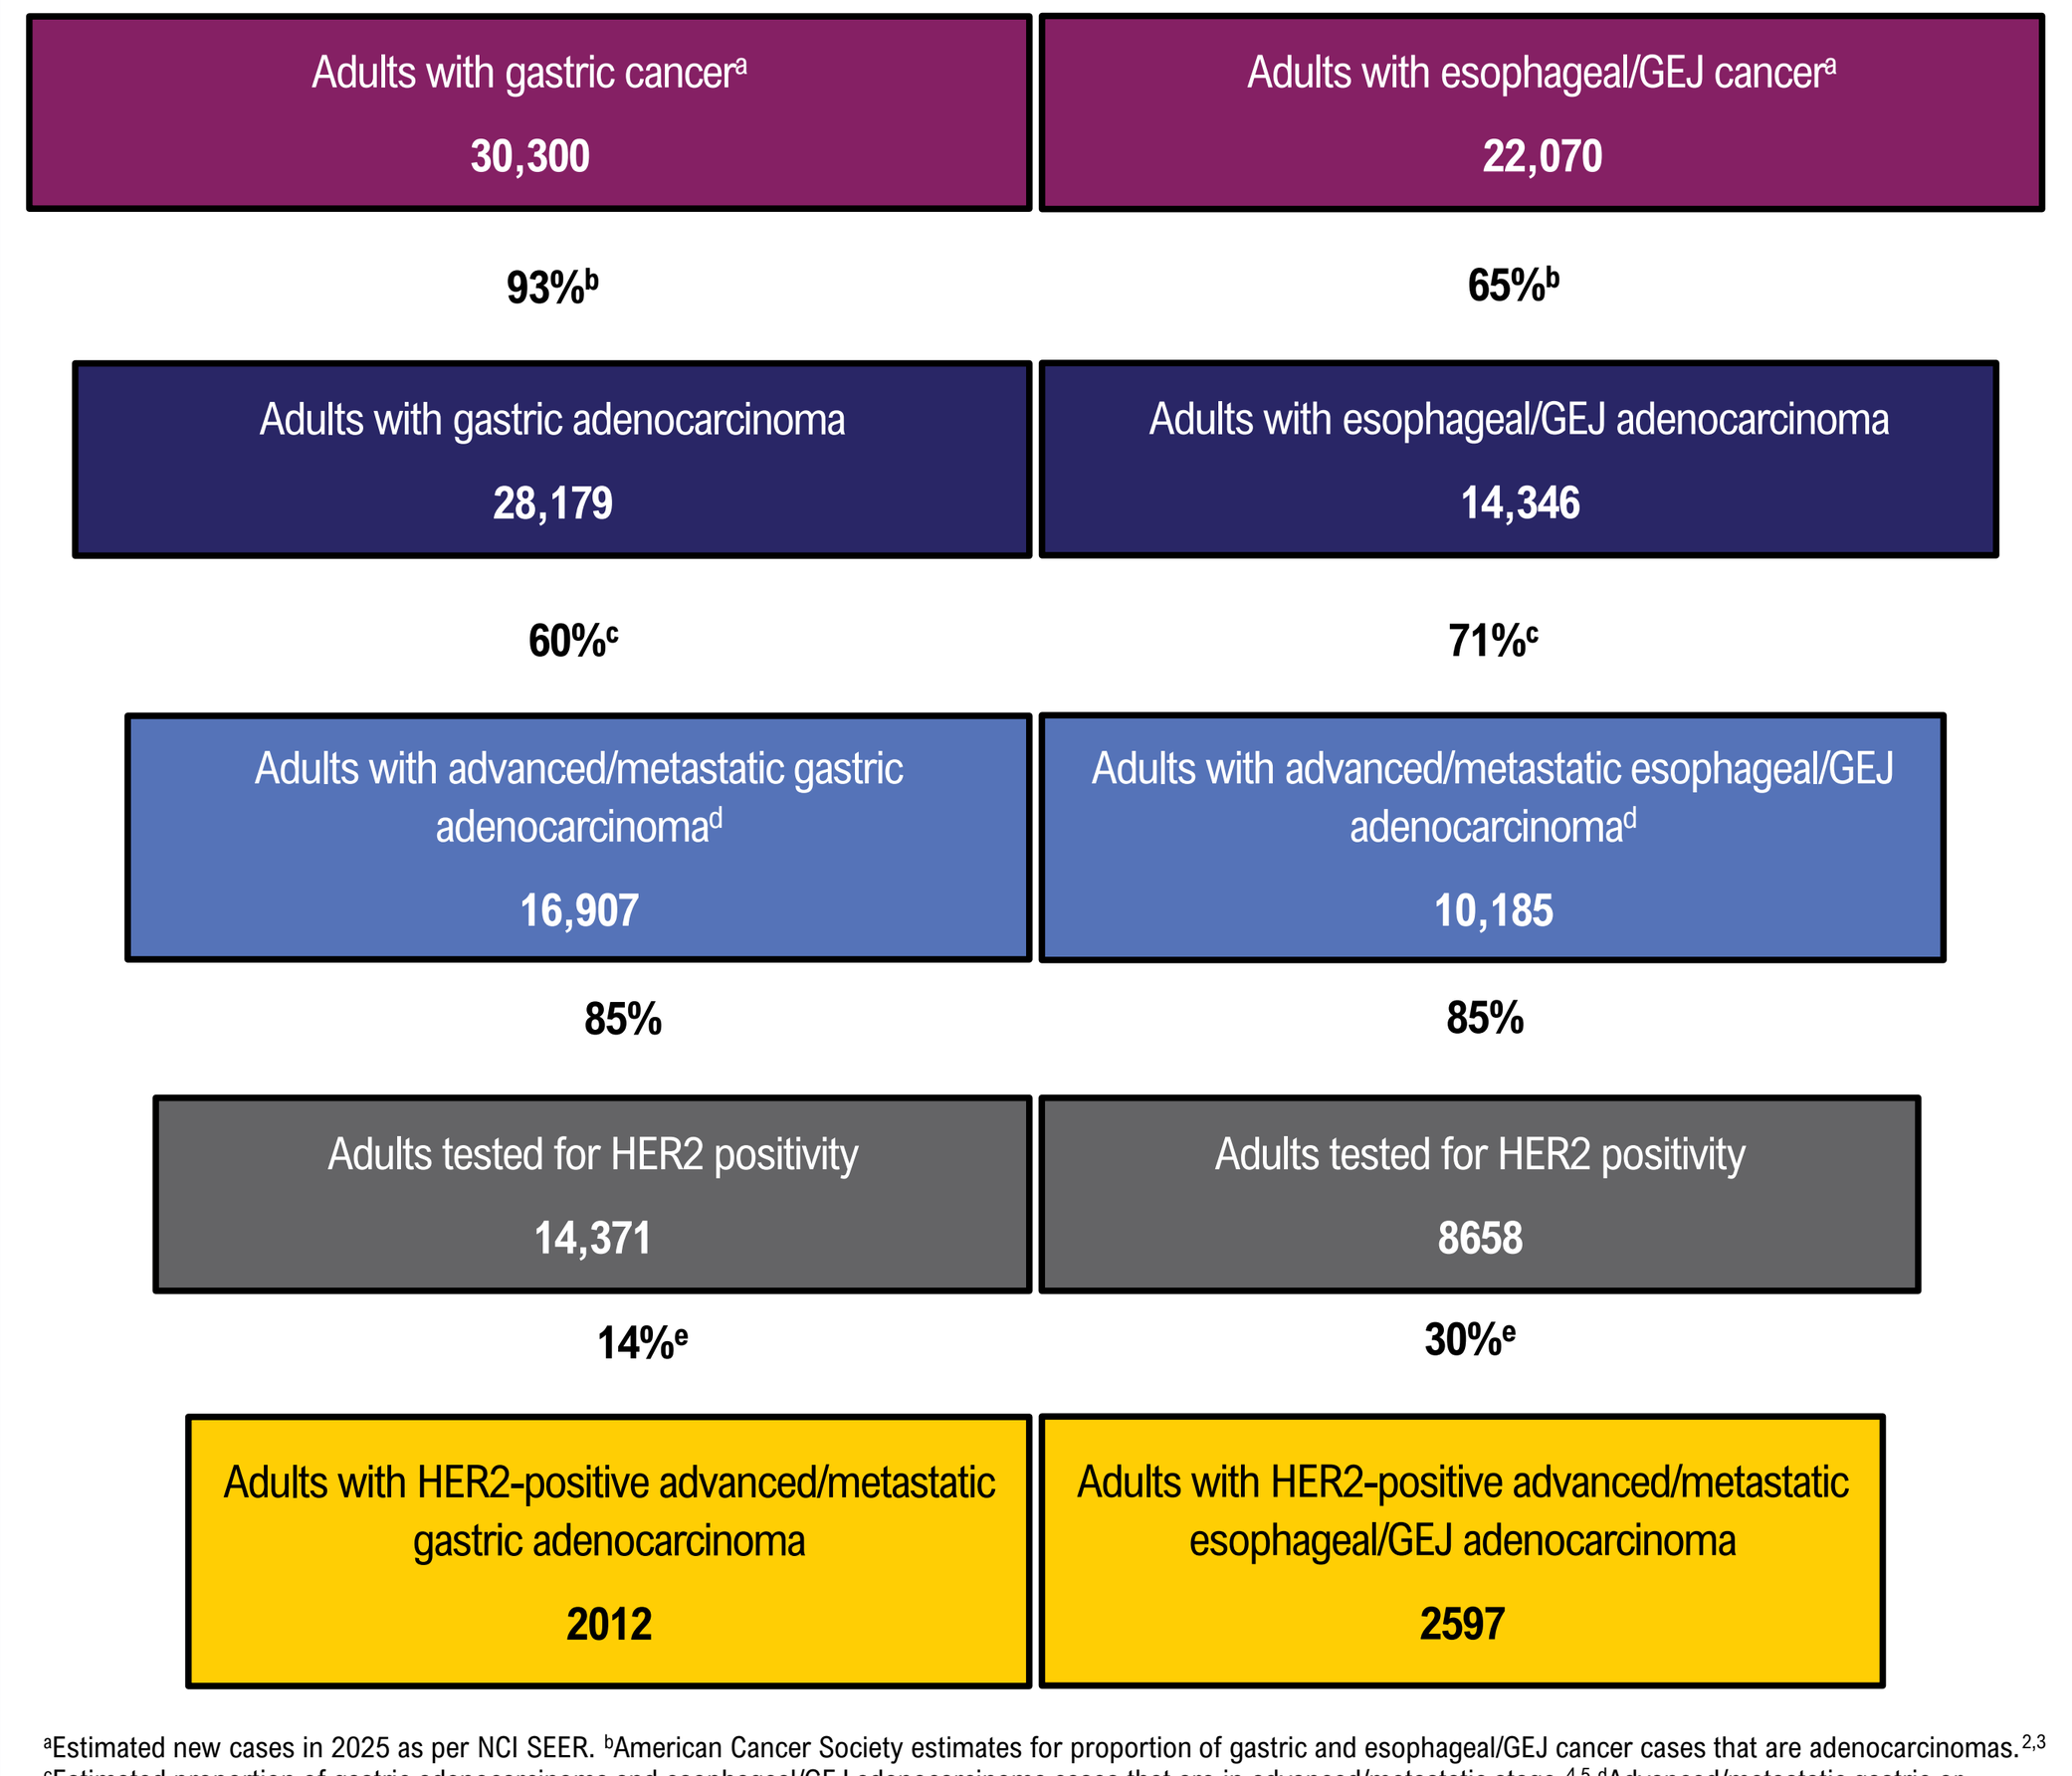

Adults with gastric cancer[a]
30,300
Adults with esophageal/GEJ cancer[a]
22,070
93%[b]
65%[b]
Adults with gastric adenocarcinoma
28,179
Adults with esophageal/GEJ adenocarcinoma
14,346
60%[c]
71%[c]
Adults with advanced/metastatic gastric adenocarcinoma[d]
16,907
Adults with advanced/metastatic esophageal/GEJ adenocarcinoma[d]
10,185
85%
85%
Adults tested for HER2 positivity
14,371
Adults tested for HER2 positivity
8658
14%[e]
30%[e]
Adults with HER2-positive advanced/metastatic gastric adenocarcinoma
2012
Adults with HER2-positive advanced/metastatic esophageal/GEJ adenocarcinoma
2597

[a]Estimated new cases in 2025 as per NCI SEER. [b]American Cancer Society estimates for proportion of gastric and esophageal/GEJ cancer cases that are adenocarcinomas.[2,3] [c]Estimated proportion of gastric adenocarcinoma and esophageal/GEJ adenocarcinoma cases that are in advanced/metastatic stage.[4,5] [d]Advanced/metastatic gastric or esophageal/GEJ adenocarcinoma includes those classified as regional or distant disease at diagnosis in the NCI SEER Program. [e]Evidence from the US Flatiron Health database for proportion of patients with advanced/metastatic GEA who are HER2-positive.[18]
GEA, gastroesophageal adenocarcinoma; GEJ, gastroesophageal junction; HER2, human epidermal growth factor receptor 2; NCI, National Cancer Institute; SEER, Surveillance, Epidemiology, and End Results Program.

- While the global epidemiology of GEA could not be accurately estimated due to lack of evidence, the weighted estimates for the US population were calculated using 2025 NCI SEER data
- Based on the calculations and data sources, the annual incidence of HER2-positive advanced/metastatic GEA in the US is estimated to be 4609 cases

Table 3. HER2 Diagnostic Outcomes Reported in Included Studies

Diagnostic Method	Biomarker Analyzed	Diagnostic Outcomes		Study Reference
		Concordance Rate	Positivity Rate	
GEA				
IHC and ISH	HER2	74%	17%	Yoon 2014[19]
NGS and IHC/ISH	HER2	57%–84%	35%	Catenacci 2023[20]; Mikhail 2015[21]
NGS	*ERBB2*	NR	15%	Zhang 2022[22]
Gastric cancer				
IHC	HER2	74%–96%[a]	8%–25%	Subasinghe 2023[23]; Kattan 2022[12]; Xu 2022[24]; Venugopalan 2021[25]; Pereira 2019[26]; Chong 2016[10]; Gupta 2016[11]
	PD-L1	NR	43%	Jing 2023[27]
ISH	HER2	NR	5%–21%	Subasinghe 2023[23]; Kattan 2022[12]; Cheng 2015[28]
IHC and ISH	HER2	50%–100%	4%–20%	Hyung 2024[29]; van der Sluis 2023[30]; Senel 2022[31]; Chen 2021[32]; Cordero-García 2019[7]; Aizawa 2014[33]
IHC and ISH vs *ERBB2* amplification (NGS)	HER2 and *ERBB2*	74%	63.3%	Hu 2023[34]
IHC 3+ vs *ERBB2* amplification (NGS)	HER2 and *ERBB2*	78%	NR	
IHC 2+ and ISH (HER2/CEP17 score ≥2) vs NGS	HER2 and *ERBB2*	79%	NR	
GC/GEJC				
IHC	PD-L1	NR	16%–41%	Hashimoto 2024[35]
ISH	*c-erb2*	NR	21%	Rajadurai 2018[15]
IHC and ISH	HER2	87%–98%	11%–37%	Hashimoto 2024[35]; Kolbe 2023[36]; Hempel 2021[37]; Seo 2017[16]; van Cutsem 2015[38]
NGS or WGS	*ERBB2*	NR	15%	Hempel 2021[37]

[a]Concordance for HER2 results using IHC was 74.2% between the 2 tissue blocks[24] and 95.8% between resection and mucosal biopsies.[25]
CEP17, centromere enumerator probe 17; GC/GEJC, gastric or gastroesophageal junction cancer; GEA, gastroesophageal adenocarcinoma; HER2, human epidermal receptor 2; IHC, immunohistochemistry; ISH, in situ hybridization; NGS, next-generation sequencing; NR, not reported; PD-L1, programmed death-ligand 1; WGS, whole-genome sequencing.

- Of the 28 diagnostic studies, 18 studies were conducted in patients with GC, 6 were in patients with GC/GEJC, and only 4 studies included patients with GEA
- Fifty-four percent of the studies evaluated HER2 using both immunohistochemistry (IHC) and in situ hybridization (ISH), including fluorescence ISH (FISH); 7 studies used IHC only, 4 studies tested IHC, ISH, and next-generation sequencing (NGS)/whole-genome sequencing, and 1 study each used FISH, NGS, and a combination of ISH with NGS
- The diagnostic criteria remained consistent across the included studies and in line with clinical guidelines.[39,40] A HER2 IHC score of 3+ was considered HER2-positive, while an IHC score of 2+ was additionally confirmed by ISH
- A high concordance rate was reported for HER2-positive status assessed using IHC and FISH in patients with GEA (74%),[19] GC (50%–100%),[7,31] and GC/GEJC (87%–98%)[36,38]
- The concordance rates for HER2-positive status in GEA assessed using NGS and IHC/FISH ranged from 57%[20] to 84%.[21] The lower rate of *ERBB2* amplification detection in one of the studies may reflect the loss of *ERBB2* amplification after progression on trastuzumab or may reflect false-negative NGS results for circulating tumor DNA due to low tumor shedding[20]
- A comparison of IHC 3+ to *ERBB2* amplification using NGS yielded a concordance of 78%, while a comparison of IHC 2+ and FISH HER2/CEP17 score ≥2 vs *ERBB2* amplification (NGS) showed a concordance of 79% in another subset of 12 patients[34]
- The HER2 positivity rate ranged from 4%[29] to 37%[36] using IHC and ISH (FISH or silver-enhanced ISH [SISH] or dual ISH) in patients with GEA, GC, or GEJC
- In a regional analysis from the comprehensive multinational ToGA study (N = 3665), HER2 positivity rates were reported as follows: 18.3% in Russia and South Africa, 24% in the Asia-Pacific region, 24% in Europe, and 16% in Central/South America[38]
- In GC, 1 study of 76 patients with HER2-positive disease found that 43% also showed programmed death-ligand 1 (PD-L1) positivity.[27] While PD-L1 positivity rates are known for GC/GEJC,[35] evidence on overlap with HER2 positivity is lacking

Limitations

- The search was limited to articles published since January 2014 to restrict it to the most recent, relevant information. It was not always possible to find information for all the research questions asked. Potentially, information on some of these topics does not yet exist in the published domain for this rare disease, or the searches need to be widened to a longer time period (ie, articles published in the last 20 years)
- Epidemiological estimates for GEA were only limited to the US, with evidence presented using Flatiron Health Oncology database[8] and by calculating annual incidence of HER2-positive advance/metastatic GEA using NCI SEER data[4,5]
- Furthermore, there was heterogeneity among the studies reporting epidemiological data in terms of study settings and sample size. Therefore, results across geographical regions should not be compared without considering these limitations

Conclusions

- This review synthesized evidence available on the epidemiology (incidence, prevalence, and mortality) of HER2 positivity in the GEA population and summarized diagnostic methods for HER2 detection, supporting improved disease understanding and future research
- Within studies that focused on epidemiological outcomes, the prevalence of HER2-positive status within the overall GEA population was 15% in the US, 14% in Europe, 11%–48% in Asia, and 4%–8% in Latin America
- Using NCI SEER, 4609 new cases of HER2-positive advanced/metastatic GEA are estimated in the US in 2025
- Among diagnostic studies included in the review, the HER2 positivity rate ranged from 4% to 37% using IHC and ISH in patients with GEA, GC, or GEJC. There was strong concordance (>70%) for HER2 diagnostic tests with IHC and ISH/FISH (excluding SISH) across multiple studies, aligning well with the diagnostic methods recommended in clinical guidelines. NGS and IHC/FISH results for the overall GEA HER2-positive population showed concordance of 57%–84%
- No evidence was identified regarding the overlap between HER2 positivity and PD-L1 expression in the overall GEA population. In GC, one study reported 43% of patients with HER2-positive disease were also PD-L1 positive, indicating a notable overlap
- Further research is needed to ascertain the epidemiology of HER2-positive GEA and its variation by anatomical sites and to determine the overlap of patients with HER2 and PD-L1 positivity

References: **1.** Petrillo A, Smyth EC, et al. *Ther Adv Med Oncol*. 2023;15:17588359231173177. **2.** American Cancer Society. 2021. Available at: https://www.cancer.org/cancer/types/stomach-cancer/about/what-is-stomach-cancer.html. Accessed: January 29, 2025. **3.** Arnold M, et al. *Gut*. 2020; 69(9):1564. **4.** National Cancer Institute. 2025. Available at: https://seer.cancer.gov/statfacts/html/stomach.html. Accessed: June 5, 2025. **5.** National Cancer Institute. 2025. Available at: https://seer.cancer.gov/statfacts/html/esoph.html. Accessed: June 5, 2025. **6.** Lau-Min KS, et al. *Clin Oncol*. 2020;38(suppl 29):12. **7.** Cordero-García E, et al. *Ecancermedicalscience*. 2019;13:962. **8.** Fleitas T, et al. *Ann Oncol*. 2023;34:S870. **9.** Gurbuz M, et al. *J Oncol Sci*. 2022; 8(1):14. **10.** Chong VH, et al. *Asian Pac J Cancer Prev*. 2016;17(5):2555-8. **11.** Gupta P, et al. *Indian J Cancer*. 2016; 53(4):505–11. **12.** Kattan J, et al. *BMC Cancer*. 2022;22(1):1114. **13.** Li Q, et al. *Cancer Med*. 2023;12(4):4110-24. **14.** Nakayama I, et al. *ESMO Open*. 2023;8(4):101582. **15.** Rajadurai P, et al. *J Gastrointest Cancer*. 2018;49(2):150-7. **16.** Seo KW, et al. *J Gastric Cancer*. 2017;17(1):52-62. **17.** Global Cancer Observatory. 2024. Available at: https://gco.iarc.who.int. Accessed: October 13, 2025. **18.** Mehta R, et al. *Curr Oncol* 2023;30(2):1869-81. **19.** Yoon HH, et al. *Cancer*. 2014;120(3):415-24. **20.** Catenacci DV, et al. *Oncology (Williston Park)*. 2023;37(4):176-83. **21.** Mikhail S, et al. *Oncotarget*. 2015;6(26):22206-13. **22.** Zhang L, et al. *JCO Precis Oncol*. 2022;6:e2100330. **23.** Subasinghe D, et al. *J Int Med Res*. 2023;51(2):3000605231154403. **24.** Xu C, et al. *Diagn Pathol*. 2022;17(1):54. **25.** Venugopalan AK, et al. *JCDR*. 2021;15(10):EC16-EC9. **26.** Pereira MA, et al. *Mol Diagn Ther*. 2019;23(6):761-71. **27.** Jing X, et al. *Cancer Med*. 2023;12(24):21905-19. **28.** Cheng Y, et al. *Clin Transl Sci*. 2015;8(4):320-5. **29.** Hyung J, et al. *Cancer Res Treat*. 2024;56(1):199-207. **30.** van der Sluis K, et al. *Histopathology*. 2023;82(6):826-36. **31.** Senel F. *J Cancer Res Ther*. 2022;18(Suppl):S299-S304. **32.** Chen L, et al. *DNA Cell Biol*. 2021;40(2):405-13. **33.** Aizawa M, et al. *Gastric Cancer*. 2014;17(1):34-42. **34.** Hu Q, et al. *Cancer Med*. 2023;12(15):16649-60. **35.** Hashimoto T, et al. *Cancer Sci*. 2024;115(5):1622-33. **36.** Kolbe K, et al. *J Cancer Res Clin Oncol*. 2023;149(3):1319-29. **37.** Hempel L, et al. *Cancers (Basel)*. 2021;13(17):4453. **38.** Van Cutsem E, et al. *Gastric Cancer*. 2015;18(3):476-84. **39.** Lordick F, et al. *Ann Oncol*. 2022;33(10):1005-20. **40.** Obermannová R, et al. *Ann Oncol*. 2022;33(10):992-1004.

Support and Acknowledgments: This analysis was funded by Jazz Pharmaceuticals. Under the direction of the authors, medical writing was provided by Lumanity (Morristown, NJ, USA) and editorial and graphic design support were provided by Red Nucleus (Yardley, PA, USA), which was funded by Jazz Pharmaceuticals, in accordance with Good Publication Practice (GPP 2022) guidelines (https://www.ismpp.org/gpp-2022). Jazz Pharmaceuticals also reviewed and edited the poster for scientific accuracy.
Disclosures: **AK** reports research funding (paid directly to his institution) from Apexigen, Astellas, BMS, Celgene, Crystal Genomics, LEAP Therapeutics, the Pancreatic Cancer Action Network (PanCAN), Parker Institute for Cancer Immunotherapy (PICI), Roche/Genentech, and Verastem; and consulting/Data and Safety Monitoring Board for Aadi, Arcus, Eisai, Fibrogen, Grail, Ipsen, Merus, and Roche/Genentech. **WS, JS,** and **JL** are employees of Jazz Pharmaceuticals, and hold stock and/or stock options in Jazz Pharmaceuticals, plc. **MS, PS,** and **AR** are employees of Lumanity. **GK** reports a consulting or advisory role with Apexigen, AstraZeneca/Daiichi Sankyo, Bristol Myers Squibb, I-Mab, Jazz Pharmaceuticals, Merck Sharp & Dohme, and Pieris Pharmaceuticals; and research funding (paid directly to his institution) from Adaptimmune, Arog, AstraZeneca/MedImmune, Bristol Myers Squibb, CARsgen Therapeutics, Daiichi Sankyo, Jazz Pharmaceuticals, Merck, Oncolys BioPharma, Pieris Pharmaceuticals, Triumvira Immunologics, Inc., and Zymeworks; travel, accommodations, and expenses from Aduro Biotech, AstraZeneca/MedImmune, Bristol Myers Squibb, ImaBiotech, Merck Sharp & Dohme, and Pieris Pharmaceuticals.

Copies of this poster obtained through QR code are for personal use only and may not be reproduced without written permission of the authors.

Corresponding author: Junji Lin
junji.lin@jazzpharma.com

Poster presented at AMCP Nexus 2025; October 27–30, 2025, National Harbor, MD, USA